Switching from a 200mg-Ritonavir (RTV, r)-Boosted Fosamprenavir (FPV) Regimen (700mg/100mg BID or 1400mg/200mg QD) to a 100mg RTV-Boosted FPV Regimen (1400mg/100mg QD) Yields Similar Efficacy and Safety (the LESS Trial)

H-1250e

C. Cohen[1], E. DeJesus[2], A. LaMarca[3], L. Sloan[4], B. Young[5], D. Sutherland-Phillips[6], L. Dix[6], L. Patel[6], C. Vavro[6], P. Wannamaker[6], M. Shaefer[6]

[1]Community Research Initiative, Boston, MA; [2]Orlando Immunology Ctr. Orlando, FL, [3]Therafirst Med. Ctr., Fort Lauderdale, FL; [4]North Texas IDC, Dallas, TX; [5]Univ of Colorado, Denver, CO; [6]GSK, RTP, NC

Abstract

Background: FPV/r 1400/100mg QD (FPVr100) has been recently approved by the FDA for use in ART-naïve patients and may offer a simplified treatment option.

Methods: Subjects on a 200mg RTV-boosted FPV regimen (FPV/r200) (vRNA <400 copies/mL [c/mL] for ≥3 months) were randomized 2:1 to FPV/r100 or continue their baseline (BL) regimen (stable NRTI background). Protocol defined virologic failure (VF) was confirmed rebound ≥400 c/mL. Primary endpoint was % of subjects who did not meet VF at or prior to Week 24 [missing/discontinuation=failure (MD=F)].

Results: BL demographics for the 209 subjects included in the ITT(E) population were: median age 44 years; 80% male; 67% white; median CD4+ cell count 434 cells/mm³. Drug-related G2-4 AEs were reported in 4% and 7% of subjects in FPV/r100 and FPV/r200 arms, respectively, most commonly diarrhea, 1% and 3%. Overall median changes in lipid parameters at Wk 24 were similar between arms with the exception of triglycerides: FPV/r100 -21 mg/dL; FPV/r200 -2 mg/dL.

Results at Wk 24	FPV/r100 N=140	FPV/r200 N=69
Subjects not meeting VF, (MD=F)*, %	92%	94%
vRNA<50 c/mL, TLOVR, %	83%	85%
vRNA<400 c/mL, TLOVR, %	92%	94%

*FPV/r100 was non-inferior to FPV/r200 at Week 24 (95% CI for treatment difference = -9.36, 5.12), p-value 0.580

Conclusions: Virologic response was similar in subjects who switched from 200mg RTV-boosted FPV to 100mg RTV-boosted FPV with few virologic failures in either arm. Adverse events were similar between groups; however subjects who switched to FPV/r100 showed greater decreases in triglyceride levels compared to those in the FPV/r200 arm.

Introduction

- When LESS was initiated, the approved once-daily (QD) dose of ritonavir (RTV, r) boosted fosamprenavir (FPV) was FPV 1400 mg + RTV 200 mg, although pharmacokinetic (PK) data suggested that a lower boosting dose of RTV (100mg QD) may be feasible.[1]
- FPV with reduced dose RTV could offer potential advantages to other regimens, including a simplified regimen (QD), greater adherence, reduced cost, and improved tolerability for patients.
- Due to limited clinical data supporting the use of a reduced dose of RTV with FPV, LESS was designed to demonstrate the non-inferiority of FPV 1400 mg + RTV 100 mg, each administered QD (FPV/r100) to a full boosted regimen of FPV + 200 mg RTV (FPV/r200).[2,3]
- FPV dosing of 1400mg + RTV 100mg QD was approved by FDA for therapy naïve patients in March, 2008.

Methods

- A phase IIIb, randomized, open-label, multicenter study evaluating the non-inferiority of a reduced dose RTV-boosted FPV regimen (FPV 1400 mg + RTV 100 mg QD [FPV/r100]) to a full RTV boosted FPV regimen (FPV 1400 mg + RTV 200 mg QD or FPV 700 mg + RTV 100 mg twice daily [BID] [FPV/r200]) in viologically suppressed subjects with a stable nucleoside reverse transcriptase inhibitor (NRTI) background receiving FPV/r200. Virologic suppression was defined as having plasma HIV-1 RNA (vRNA) levels <400 copies/mL (c/mL) for at least 3 months prior to and at screening.

Methods

- Subjects were eligible if they were appropriate candidates for a reduced-RTV boosting dose FPV regimen (i.e. had been on no more than one antiretroviral [ART] regimen prior to switching to FPV/r200, and they must not have experienced virologic failure [VF] or any viral rebound [vRNA ≥2000 c/mL] on the prior regimen).
- Two-level stratification occurred prior to randomization according baseline (BL) FPV/r regimen (either FPV/r 1400mg/200mg QD or FPV/r 700mg/100mg BID) and prior ART (no previous PI use, non-boosted PI use, or boosted PI use).
- The primary endpoint was the proportion of subjects who did not meet virologic failure by Week 24 (missing/discontinuation=failure [MD=F]). Virologic failure was defined as two consecutive plasma vRNA levels ≥400 c/mL.
 - Secondary endpoints included proportion of subjects with vRNA <50 c/mL and <400 c/mL at Week 24, immunologic changes, virology, safety, and pharmacokinetics (steady-state amprenavir [APV] and RTV Ctau).

Figure 1. Study Design

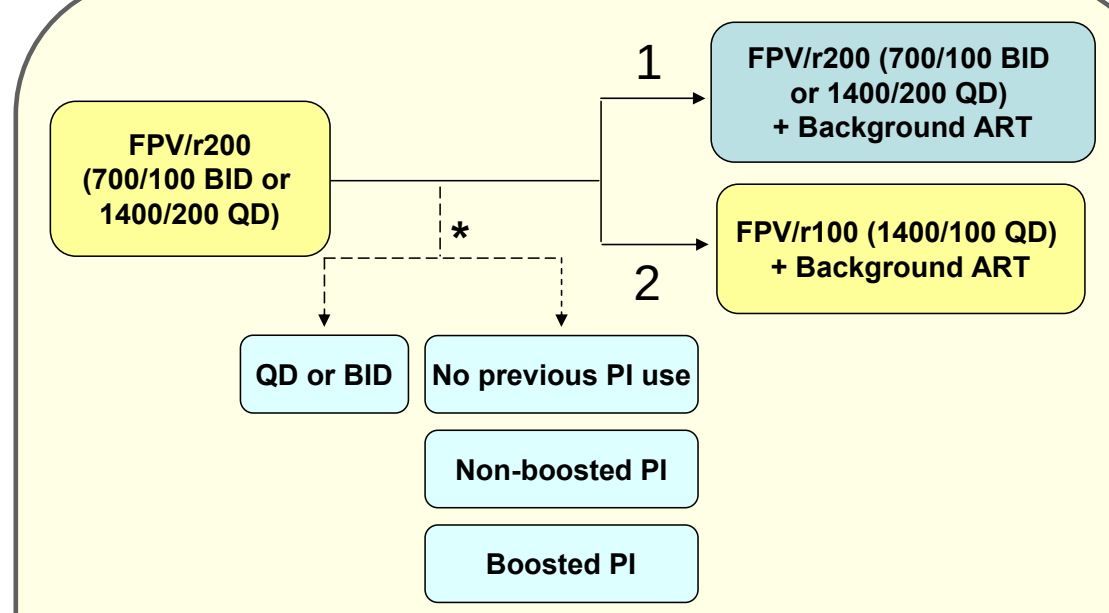

FPV/r200 (700/100 BID or 1400/200 QD)
1
FPV/r200 (700/100 BID or 1400/200 QD) + Background ART
2
FPV/r100 (1400/100 QD) + Background ART
*
QD or BID
No previous PI use
Non-boosted PI
Boosted PI

*Randomization

NOTE: The original sample size of 348 subjects was reduced to 211 due to slow enrollment; sample size re-estimation was conducted and the study retained 90% power for the primary endpoint after the reduction in sample size.

Results

- A total of 210 subjects were enrolled; 209 subjects were included in the ITT-E population.
- Almost half of all subjects were on abacavir (ABC)/lamivudine (3TC) as their NRTI backbone (FPV/r100: 49%; FPV/r200: 41%) followed by tenofovir (TDF)/emtricitabine (FTC) (FPV/r100: 20%; FPV/r200: 29%); 3TC/zidovudine (ZDV) (FPV/r100: 20%; FPV/r200: 17%); and Other (FPV/r100: 11%; FPV/r200: 13%).

Acknowledgements

The authors would like to extend their thanks to the subjects who participated in this trial and to the study investigators, study coordinators, and the GSK study team for their invaluable contributions.

References

1. Ruane, P. et al. 2004 ICAAC, Washington DC, USA. Abstract A-449.
2. Smith, K. et al. 2007 IAS, Sydney, AU. Abstract WEPEB023.
3. Hicks, C. et al. 2007 11th European AIDS Conference, Madrid, SP. Abstract P5.7/01.

Table 1. Baseline Characteristics, ITT-E Population

	FPV/r100 N=140 n (%)	FPV/r200 N=69 n (%)
Mean age, years	45	44
Male	107 (76%)	60 (87%)
Race/Ethnicity		
White/Caucasian	95 (68%)	46 (67%)
African American	42 (30%)	20 (29%)
Other	2%	4%
Hispanic or Latino	31 (22%)	11 (16%)
Hepatitis Status		
Hepatitis B positive	3 (2%)	2 (3%)
Hepatitis C positive	13 (9%)	11 (16%)
% with BL HIV-1 RNA <400 c/mL	138 (99%)	69 (100%)
BL median CD4+ cell count (cells/mm³)	432	438
CDC Class C	29 (21%)	17 (25%)

Table 2. Subject Disposition, ITT-E Population

	FPV/r100 N=140 n (%)	FPV/r200 N=69 n (%)
Completed	133 (95%)	66 (96%)
Prematurely Withdrawn	7 (5%)	3 (4%)
Primary Reason for Withdrawal		
Subject Decision	3 (2%)	1 (1%)
Adverse Event	2 (1%)	0
Noncompliance	1 (<1%)	1 (1%)
Lost to Follow Up	1 (<1%)	0
Protocol Defined Virologic Failure	0	1 (1%)

Table 3. Proportion of Subjects Without Failure, MD=F, ITT-E Population

	FPV/r100 n/N (%)	FPV/r200 n/N (%)
Proportion Without Failure[1]	92%	94%
Response by baseline FPV/r dose, n/N (%)		
FPV/r 700mg/100mg BID	64/68 (94%)	35/37 (95%)
FPV/r 1400mg/200mg QD	65/72 (90%)	30/32 (94%)
Response by previous PI experience, n/N (%)		
None	55/60(92%)	28/30 (93%)
Non-boosted PI	27/28 (96%)	12/12 (100%)
Boosted PI	47/52 (90%)	25/27 (93%)

1. 95 CI for treatment difference -2.12 (-9.36, 5.12), p value 0.580.

- Similar proportions of subjects (FPV/r100: 92% and FPV/r200: 94%) in both treatment arms were without failure at Week 24. The lower bound of the 95% confidence interval for the difference in proportions between treatment groups (FPV/r100 minus FPV/r200) was greater than the predefined delta of -0.12; therefore, non-inferiority of FPV/r100 compared with FPV/r200 was established.
- Median CD4+ cell counts at Week 24 were 446 cells/mm³ and 438 cells/mm³ for the FPV/r100 and FPV/r200 group, respectively.

Protocol Defined Virologic Failure

- Only one subject (FPV/r200 arm) experienced protocol defined virologic failure during the 24 week study period. No major PI mutations emerged on treatment nor was there any reduced drug susceptibility in this subject.

Figure 2. Proportion of Subjects with Plasma HIV-1 RNA <50 c/mL and <400 c/mL by Week 24, ITT-E Population

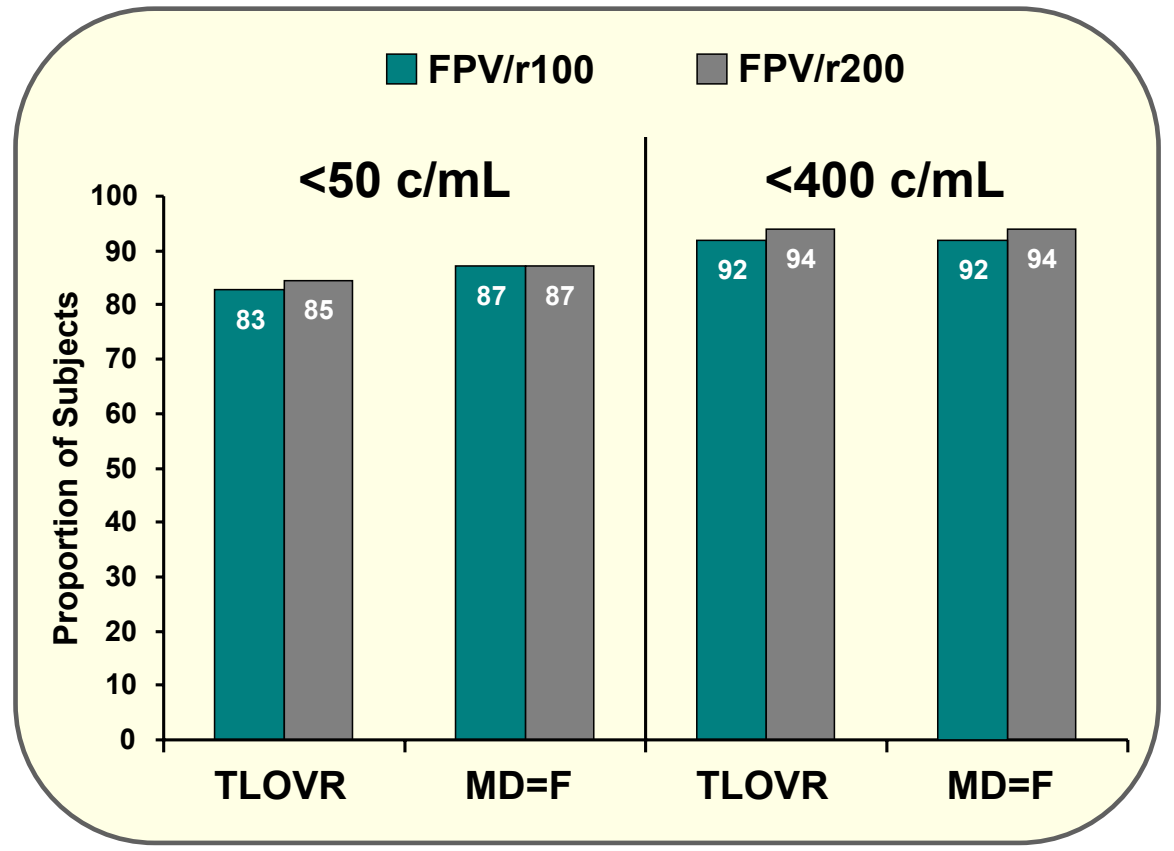

FPV/r100
FPV/r200
<50 c/mL
<400 c/mL
Proportion of Subjects
100
90
80
70
60
50
40
30
20
10
0
83
85
87
87
92
94
92
94
TLOVR
MD=F
TLOVR
MD=F

Table 4. Safety Results, Safety Population

	FPV/r100 N=142 n (%)	FPV/r200 N=67 n (%)
Any Grade 2-4 Adverse Event (AE)	48 (34%)	24 (36%)
Treatment-Related Grade 2-4 AEs (≥3% in either arm)	5 (4%)	5 (7%)
Diarrhea	2 (1%)	2 (3%)
Hypercholesterolemia	0	2 (3%)
Any Serious Adverse Event (SAE)	7 (5%)	1 (1%)
Treatment-Related SAEs	0	0

*One death occurred during the study period (FPV/r100 arm) which was not considered treatment-related by the investigator.

Table 5. Median Changes in Fasting Lipid Parameters (mg/dL)

	FPV/r100			FPV/r200		
	BL mg/dL	Wk 24 mg/dL	Δ from BL	BL mg/dL	Wk 24 mg/dlL	Δ from BL
Total Cholesterol	203	197	-1	197	200	1.5
LDL Cholesterol	115	116	0	103	106	2.5
HDL Cholesterol	48	48	-1	48	47	0
Triglycerides	172	152	-21	195	180	-1

- Minimal changes in fasting lipid parameters were noted for both treatment groups although subjects in the FPV/r100 group did appear to demonstrate greater decreases in triglyceride levels by Week 24 (FPV/r100: -21 mg/dL; FPV/r200: -1 mg/dL).

Pharmacokinetics

- Average mean plasma amprenavir (APV) Ctau levels were 1.84 mcg/mL (range 0.230 to 9.935 mcg/mL), 1.74 mcg/mL (range 0.293 to 2.997 mcg/mL), and 2.3 mcg/mL (range 0.010 to 4.920 mcg/mL) for subjects who switched to FPV/r 1400mg/100mg QD (n=61), remained on FPV/r 1400mg/200mg QD (n=17), or remained on FPV/r 700mg/100mg BID (n=17), respectively. These concentrations are well above the historical mean APV protein binding (90%) adjusted IC_{50} for wild-type HIV of 0.146 mcg/mL.[1]

Discussion

- In this study, a population of subjects who were on a stable and virologically suppressive regimen (vRNA <400 c/mL for ≥3 months) and were switched to a reduced dose of RTV boosted FPV (FPV/r100) continued to maintain virologic suppression through 24 weeks of study.
 - The proportion of subjects who were not failures at Week 24 was similar regardless of their baseline regimen (FPV/r200 QD or BID) or previous PI use (no previous PI use, unboosted PI, or boosted PI). However, subjects with previous virologic failure or virologic rebound were excluded from this study.
- Grade 2-4 adverse events were experienced by approximately 34% of subjects; most were not considered related to treatment. This finding is not surprising since all subjects were stable on an FPV/r200 regimen prior to study entry. These results may not represent perceived tolerability differences in these regimens in other study populations, e.g ART-naïve.[2,3]

Conclusions

- A regimen of FPV 1400 mg + RTV 100 mg demonstrated non-inferiority to full boosted FPV/RTV (700/100 mg BID or 1400/200 mg QD) over a 24 week period in virologically stable subjects.
- Only 1 subject experienced protocol-defined virologic failure.
- Overall, both regimens were generally well tolerated and no subject experienced a treatment-related SAE.
- Lipid changes were similar between the treatment groups
 - Subjects in the FPV/r100 arm experienced greater decreases in triglycerides (-21 mg/dL) compared to the FPV/r200 arm (-1 mg/dL).

48th Annual Interscience Conference on Antimicrobial Agents and Chemotherapy Meeting, 25–28 October 2008, Washington, DC, USA