WEPEA097

S/GSK1349572 is a Potent Next Generation HIV Integrase Inhibitor

Akihiko Sato[1], Masanori Kobayashi[1], Tomokazu Yoshinaga[1], Tamio Fujiwara[1]
Mark Underwood[2], Brian Johns[2], Scott Foster[2], Richard Hazen[2],
Robert Ferris[2], Kevin Brown[2], Edward Garvey[2]
[1]Shionogi & Co., Ltd., Osaka, Japan ; [2] GlaxoSmithKline, Research Triangle Park, NC, US.

SHIONOGI gsk GlaxoSmithKline
Shionogi-GlaxoSmithKline Pharmaceuticals, LLC

Abstract

Background: S/GSK1349572 was selected as lead molecule from a series of two-metal binding integrase inhibitor scaffolds. We report here its antiviral potency, mechanism of action, and resistance profile.

Methods: Antiviral activity of S/GSK1349572 was measured against different HIV strains in various cell lines. Mechanism of action was determined using *in vitro* integrase strand transfer and binding assays, effects on viral DNA integration, and resistance passage experiments. Fold change (FC) in susceptibility against integrase inhibitor (INI)-resistant mutants was determined with a HeLa-CD4 cell line assay.

Results: S/GSK1349572 inhibited HIV integrase in an *in vitro* strand transfer assay. In PBMC cells, the antiviral EC50 and EC90 values were 0.51 and 2.0 nM, respectively. In MT-4 antiviral assays, the potency shift extrapolated to 100% human serum was 75-fold, resulting in estimated PA-EC90 of 152 nM in PBMCs. S/GSK1349572 had low nM potency against a broad clade panel of HIV-1 isolates representing Groups M and O, and against HIV-2. The mean EC50 against HIV-1 from thirteen subtype B clinical isolates was 0.52 nM. In cellular assays, S/GSK1349572 blocked viral DNA integration with a concomitant increase in 2-LTR circles. When virus was passaged in the presence of S/GSK1349572, highly resistant mutants were not isolated, but mutations which conferred low FC (maximum FC=4.1) were identified within the integrase active site. S/GSK1349572 demonstrated low fold change in activity against raltegravir resistant site directed molecular clones, including Y143R (S/GSK1349572 FC=1.4 and RAL FC=16), Q148K (S/GSK1349572 FC=1.1 and RAL FC=83), N155H (S/GSK1349572 FC=1.2 and RAL FC=11), and G140S, Q148H (S/GSK1349572 FC=2.6 and RAL FC=>130).

Conclusions: S/GSK1349572 was a potent inhibitor of HIV integrase *in vitro* and in cellular HIV replication assays. S/GSK1349572 had a markedly different resistance profile as evidenced by limited cross-resistance to raltegravir resistant molecular clones and the low level maximum resistance to S/GSK1349572 in serial passage.

Introduction

- The long-standing Shionogi-GSK Joint Venture has made considerable progress in developing next-generation integrase inhibitors.
- S/GSK1349572 is the only once-daily, unboosted integrase inhibitor currently in development with unprecedented antiviral activity and a superior resistance profile.[1,2,3]
- S/GSK1349572 has demonstrated a predictable, well-characterized exposure-response relationship and low PK variability.[3]

Methods

In vitro strand transfer assay; Recombinant HIV integrase was used in an *in vitro* strand transfer assay which measured the amount of donor DNA covalently attached to target DNA.

Cell based antiviral & cytotoxicity assays; Antiviral & cytotoxicity assays were performed using several virus strains and multiple cell lines. Testing of S/GSK1349572 against various HIV clades and HIV-1 clinical isolates was done at Southern Research Institute in PBMCs and at Monogram Biosciences using their Integrase Phenosense assay. The effects of protein binding on antiviral activity were tested using human serum albumin or human serum.

Analysis of antiviral mechanism; Mechanistic studies examined the effect of compound on viral DNA production and integration using quantitative PCR.[4]

Passage study; Generation of virus resistant to inhibitor was performed by passage of IIIB virus in MT2 cells in the presence of S/GSK1349572. For comparison, the potency of INIs that have been studied in the clinic (raltegravir and elvitegravir) were also measured in the above assays. All compounds were synthesized either at GSK or Shionogi.[5,6]

Results and Discussion

Table 1. Inhibition of Recombinant HIV Integrase and HIV Replication by S/GSK1349572

	Integrase, nM	Antiviral, nM			
INI	Strand transfer IC_{50}	PBMC IC_{50}	Potency Shift with 100% HS	PA IC_{50}	PA IC_{90}
S/GSK1349572	2.7	0.51	75	38	152

- S/GSK1349572 inhibited recombinant HIV integrase in a strand transfer assay with nanomolar potency.
- The effective IC_{50} was determined by accounting for the antiviral IC_{50} fold shift in presence of protein. *In vitro* antiviral assays in the presence of increasing human serum were used to calculate the 75-fold shift in IC_{50} value of S/GSK1349572 in 100% human serum (by method of extrapolation); the protein-adjusted IC_{50} (PA IC_{50}) for S/GSK1349572 is estimated to be 38 nM.
- S/GSK1349572 was tested for cytotoxic effects in IM-9, U-937, MT-4, Molt-4 and PBMCs (stimulated and non-stimulated), with $CCIC_{50}$ values of 4.8, 7.0, 14, 15, 52 and 189 μM, respectively.

Table 2. Inhibition of Different HIV Clades and Clinical Isolates by S/GSK1349572

Assay	N	Mean IC_{50} (nM) [range]
HIV-1 Isolates in PBMCs	25	0.36 [0.02-2.14]
HIV-1 Isolates in MDMs	3	0.87 [0.37-1.98]
HIV-2 Isolates in PBMCs	3	1.07 [0.09-0.61]
HIV-1 containing Integrase coding region from clade B isolates	13	0.52 [0.41-0.60]
HIV1 IIIB	1	0.46
HXB2	1	0.65

- Isolates examined in PBMCs included 3 each of clades A-G, and group O. There were 16 CCR5 tropic, 5 CXCR4 tropic, and 1 undetermined tropic HIV-1.
- S/GSK1349572 was determined to be highly active with broad activity against a total of 26 unique HIV−1 isolates and three HIV-2 isolates.
- S/GSK1349572 was also active against HIV-1 containing the integrase coding region from 13 clade B clinical isolates and two control lab strains (HIV−1 IIIB and HXB2) in an isogenic background.

Figure 1. S/GSK1349572 Inhibits Integration of Viral DNA with A Concomitant Increase in 2-LTR Circles, with No Effect on Viral DNA Production

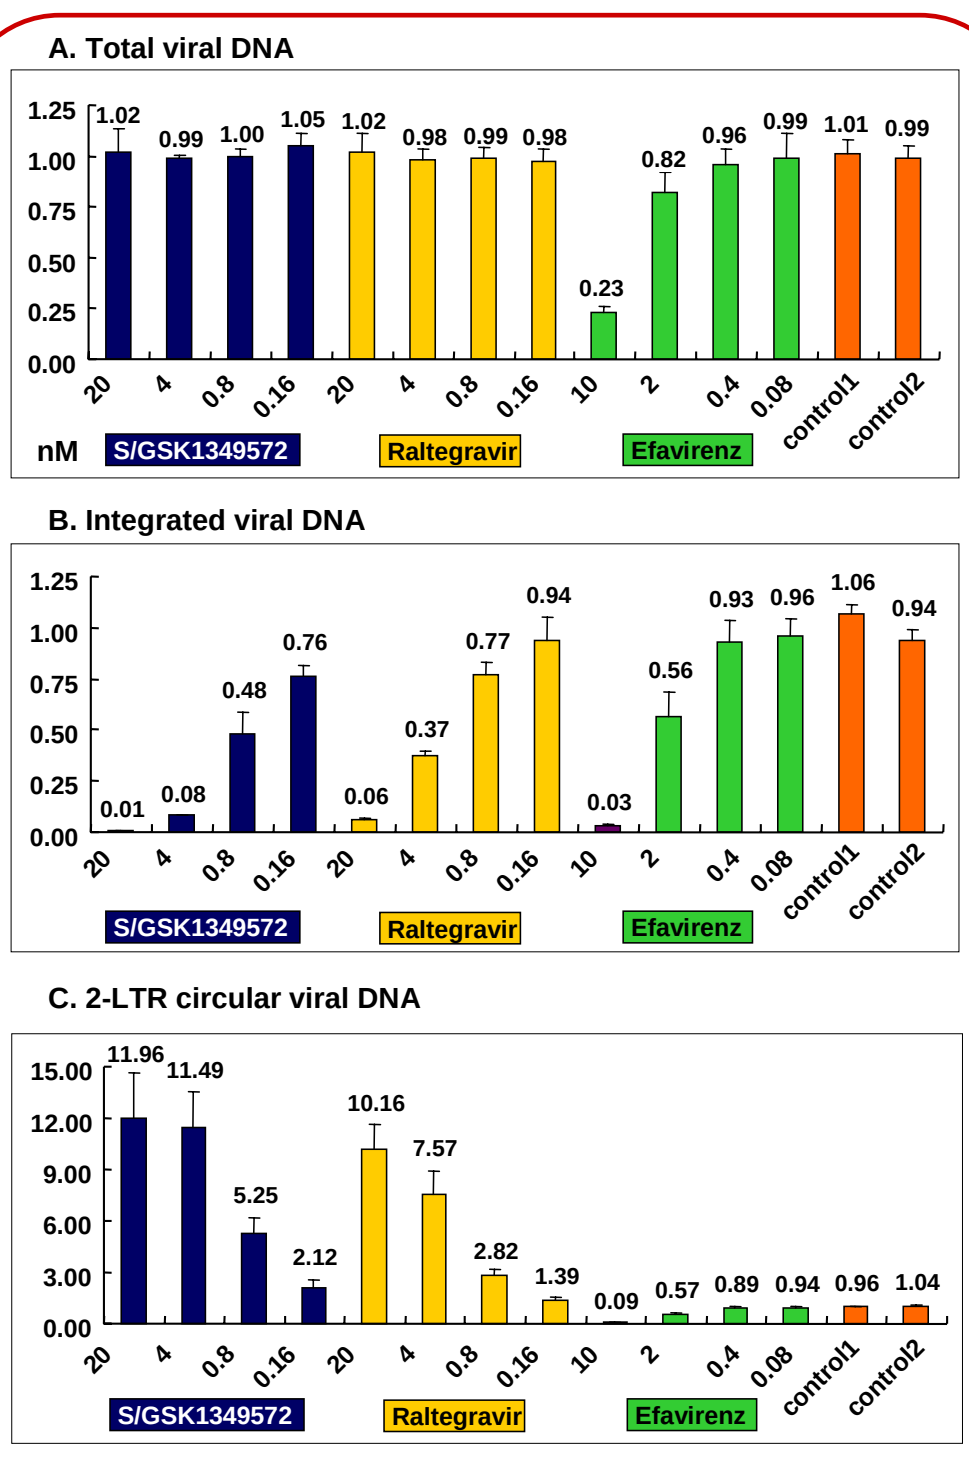
A. Total viral DNA
1.25
1.00
0.75
0.50
0.25
0.00
1.02
0.99
1.00
1.05
1.02
0.98
0.99
0.98
0.23
0.82
0.96
0.99
1.01
0.99
20
4
0.8
0.16
20
4
0.8
0.16
10
2
0.4
0.08
control1
control2
nM
S/GSK1349572
Raltegravir
Efavirenz
B. Integrated viral DNA
0.01
0.08
0.48
0.76
0.06
0.37
0.77
0.94
0.03
0.56
0.93
0.96
1.06
0.94
C. 2-LTR circular viral DNA
15.00
12.00
9.00
6.00
3.00
0.00
11.96
11.49
5.25
2.12
10.16
7.57
2.82
1.39
0.09
0.57
0.89
0.94
0.96
1.04

X axis: Concentration of antiviral in nM. Control 1 & 2 were without antiviral.
Y axis: Relative amount of DNA vs. mean of DNA amount of control 1 & 2

- Although S/GSK1349572 did not affect the amount of late RT products, S/GSK1349572 inhibited integration of HIV cDNA in dose dependent manner.
- Both S/GSK1349572 and RAL increased the amount of 2-LTR circles, while EFV decreased it.

Results and Discussion (Cont)

Figure 2. *In vitro* Passage Selected Virus Was Able to Replicate under Higher Raltegravir (RAL) Concentrations than for S/GSK1349572 (572)

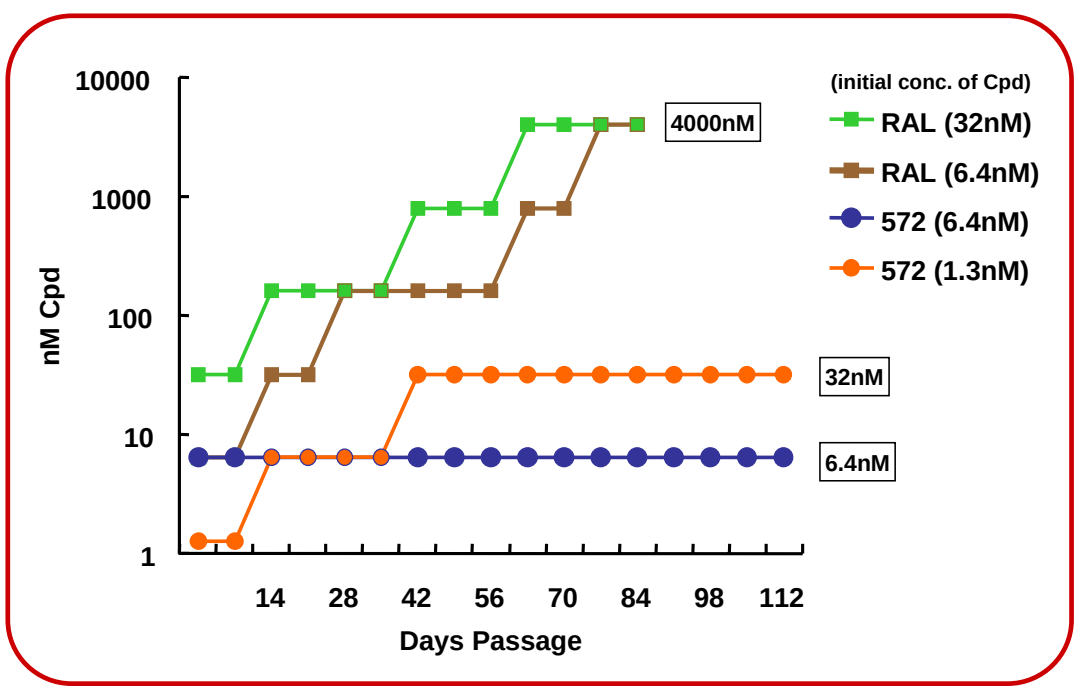
10000
1000
100
10
1
nM Cpd
14
28
42
56
70
84
98
112
Days Passage
(initial conc. of Cpd)
RAL (32nM)
RAL (6.4nM)
572 (6.4nM)
572 (1.3nM)
4000nM
32nM
6.4nM

- HIV-1 IIIB was passaged in medium containing increasing concentrations of S/GSK1349572 or RAL.
- Viruses with RAL-resistant mutation(s) replicated in the presence of RAL at the higher concentration of 4,000nM.
- Genotypic assay identified that N155H, Q148K, or Q148R were selected during passages with RAL by day 28 or 42, and resulted in phenotypic resistance (FC=19, >23, or 8) against RAL.
- HIV was unable to replicate under an initial concentration of 32nM S/GSK1349572, and no replication was observed under 160nM during passage.
- These *in vitro* passage data demonstrate the potential for a higher genetic barrier for S/GSK1349572 when compared to RAL.

Table 3. Integrase Mutations Generated by Passage of Virus in the Presence of S/GSK1349572, Raltegravir (RAL), or Elvitegravir (ELV)

Raltegravir (84 days)	Elvitegravir (56 days)
T124A	T66I
Q148K*	E92Q
Q148R	T124A
E138K/Q148K	P145S
E138K/Q148R	Q148K
G140S/Q148R	Q148R
N17S/Q148K/G163R	T66I/T124A
G140C/Q148K/G163R	T66K/T124A
E138K/Q148K/G163R	E92V/T124A
E92Q/E138K/Q148K/M154I	P145S/T124A
N155H/I204T	Q146L/T124A
V151I/N155H	Q148R/T124A
T124A/V151I/N155H	T66I/V72A/A128T
	T66I/E92Q/T124A
	T66I/T124A/Q146L
(FC=6 - >138)	(FC=2 - 497)

S/GSK1349572 (56 days)	S/GSK1349572 (84 days)	S/GSK1349572 (112 days)
T124A	T124A	T124A
T124A/S153F	S153Y	S153Y
	T124A/S153Y	T124A/S153Y
	L101I/T124A/S153F	L101I/T124A/S153F
		(FC=1.2 – 4.1)

Red text indicates substitutions seen in clinical trials

- T124A is polymorphic and has wild type potency versus site directed T124A mutants.
- Highly resistant mutants with high fold change (FC>100) were isolated in the presence of RAL and ELV; many of these mutations have been observed in the clinic in patients failing RAL and ELV-based regimens.
- In the presence of S/GSK1349572, highly resistant mutants were not isolated. Multiple mutations in INI selected during S/GSK1349572 passage only conferred low fold change (maximum FC=4.1).
- Data may be consistent with higher genetic barrier of S/GSK1349572 when compared to RAL and ELV.

Figure 3. Susceptibility of INI Resistant Molecular Clones to S/GSK1349572

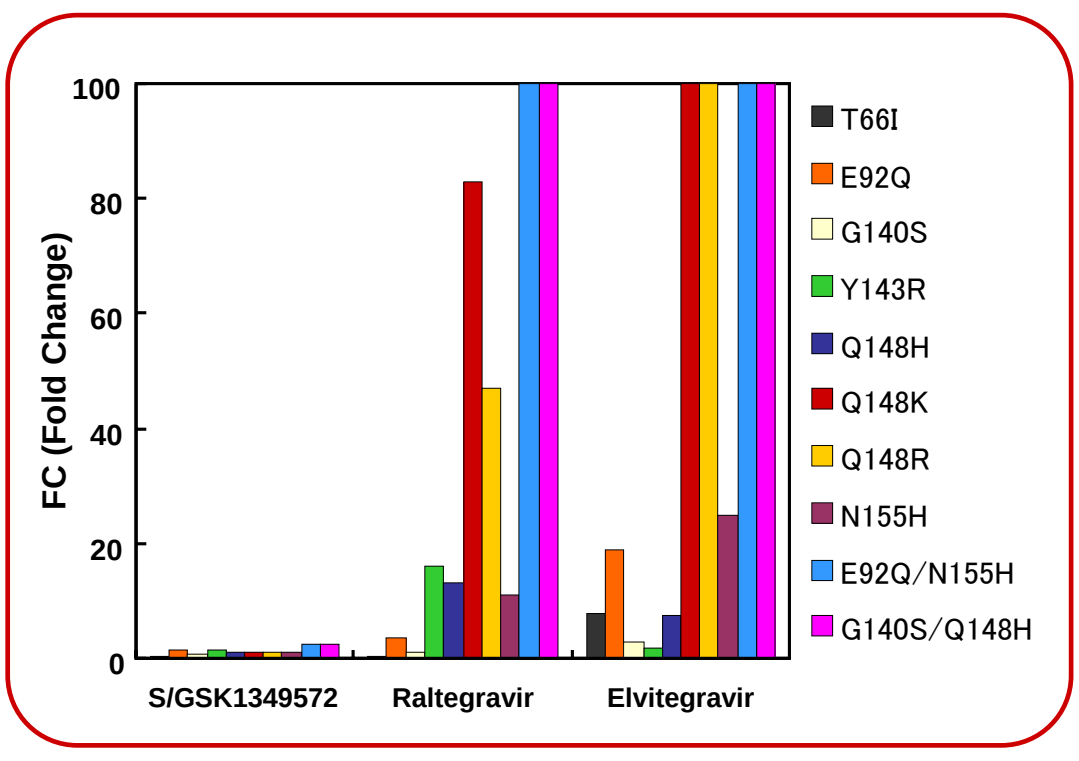
100
80
60
40
20
0
FC (Fold Change)
S/GSK1349572
Raltegravir
Elvitegravir
T66I
E92Q
G140S
Y143R
Q148H
Q148K
Q148R
N155H
E92Q/N155H
G140S/Q148H

- INI-resistant mutant HIV-1 viruses were produced from wild type virus NL-432 using site directed mutagenesis of the integrase coding region and were tested for susceptibility to INIs using infected HeLa-CD4 cells carrying a reporter β-galactosidase gene driven by HIV-1 LTR.
- Fold change (FC) of each clone was determined in comparison with the IC_{50} value for the wild type virus NL432.
- S/GSK1349572 demonstrated low fold change in activity against RAL and ELV-resistant site directed molecular clones.
- Although limited cross-resistance between RAL and ELV was observed, S/GSK1349572 was active against this panel of INI-resistant mutants.

Conclusions

- S/GSK1349572 is a potent inhibitor of HIV integrase *in vitro* and in cell based HIV replication assays.
- The S/GSK1349572 mechanism of action was confirmed by measuring HIV integrase strand transfer inhibition in biochemical assay, by viral DNA species quantification after viral infection in cells in the presence and absence of antivirals, and by passaging HIV-1 to select resistance followed by phenotypic and genotypic analysis.
- S/GSK1349572 inhibited HIV replication in a clade- and clinical isolate-independent fashion.
- S/GSK1349572 had limited cross-resistance to RAL- and ELV-resistant mutants.
- *In vitro* passage study showed that S/GSK1349572 leads to a less diverse resistance profile with lower fold change.
- *In vitro* experiments support the potential for S/GSK1349572 to have a higher genetic barrier to resistance when compared to 1st generation INIs.

Acknowledgements

The discovery and pre-clinical development of S/GSK1349572 was the product of a Shionogi-GSK research collaboration and resulted from the talent and devotion of the entire HIV integrase team at both companies.

References

1. Lalezari J. et al. IAS 2009, Cape Town, Oral #TUAB105.
2. Underwood M. et al. IAS 2009, Cape Town, Poster #WEPEA098.
3. Song I, et al. IAS 2009, Cape Town, Poster #WEPEB250.
4. Garvey EP et al. AAC. 2008; 52: 901-08.
5. Kobayashi M et al. Antiviral Research. 2008; 80: 213-22.
6. Nakahara K et al. Antiviral Research. 2008; 81: 141-46.

IAS 2009- 5th Conference on HIV Pathogenesis, Treatment and Prevention 19-22 July 2009, Cape Town South Africa