Poster
MOPEB033

Differences in Virologic Response Among African-Americans and Females Regardless of Therapy in the HEAT Study

Smith KY[1], Kumar PN[2], Patel P[3], Bellos NC[4], Sloan L[5], Lackey P[6], Sutherland-Phillips DH[3], Vavro C[3], Yau L[3], Shaefer MS[3]
[1]Rush University Medical Center, Chicago, IL USA; [2]Georgetown University, Washington, DC, USA; [3]GlaxoSmithKline, Research Triangle Park, NC, USA; [4]Southwest Infectious Disease Associates, Dallas, TX, USA; [5]North Texas Infectious Disease Consultants, Dallas, TX, USA; [6]ID Consultants, Charlotte, NC, USA

Abstract

Objectives: **Differences in virologic and immunologic response have been observed with different antiretroviral combinations. We evaluated week 96 responses by sex and by race in the HEAT study which previously demonstrated the non-inferiority of abacavir sulfate/lamivudine (ABC/3TC) versus tenofovir/emtricitabine (TDF/FTC), when each was combined with lopinavir/ritonavir (LPV/r), at 48 weeks and subsequently confirmed at 96 weeks in ART-naïve subjects.**

Methods: **Virologic response at week 96 was determined by the proportion with HIV-1 RNA (VL) <50 c/mL by missing=failure (M=F) and observed analyses. Race and ethnicity were self-identified.**

Results: **Immunologic and Virologic Response at Week 96**

	ABC/3TC + LPV/r N=343			TDF/FTC + LPV/r N=345		
	Median CD4+ Count BL (Δ96)	VL<50 c/mL Obs % (n/N)	VL <50 c/mL M=F % (n/N)	Median CD4+ Count BL (Δ96)	VL<50 c/mL Obs % (n/N)	VL <50 c/mL M=F % (n/N)
All Subjects	214 (+250)	87% (205/236)	60% (205/343)	193 (+247)	91% (200/219)	58% (200/345)
Female	213 (+307)	84% (179/205)	46% (26/56)	168 (+262)	90% (36/40)	52% (36/69)
Male	214 (+245)	87% (179/205)	62% (179/287)	200 (+245)	92% (164/179)	59% (164/276)
African-American*	182 (+245)	80% (60/75)	50% (60/119)	137 (+245)	86% (59/69)	48% (59/124)
Caucasian	209 (+225)	89% (97/109)	68% (97/143)	237 (+255)	95% (99/104)	67% (99/147)
Hispanic	247 (+323)	93% (42/45)	58% (42/73)	179 (+236)	92% (36/39)	58% (36/62)
Other	184 (+249)	86% (6/7)	75% (6/8)	232 (+151)	86% (6/7)	50% (6/12)

*African-American race, but not female sex, was a significant predictor of virologic failure in a multivariate logistic regression model studied.

Conclusions: **Differences in CD4+ and virologic responses by sex and race at week 96 were observed. Notably, fewer African-Americans and females achieved VL <50 c/mL in both arms and in both analyses suggesting these differences may be multifactorial. Slightly greater CD4+ increases were observed in females and although African-Americans had lower initial CD4 counts, immunologic recovery was similar across other racial groups and between groups over 96 weeks. Further investigation into the underlying differences in response is required.**

Introduction

- **Differences in response to antiretroviral therapy appear to be associated with numerous factors including sex and subgroups of race/ethnicity. [1]**
- **HEAT, a randomized, double-blind, placebo-matched, multicenter trial demonstrated the non-inferiority of abacavir sulfate/lamivudine (ABC/3TC) to tenofovir/emtricitabine (TDF/FTC), each in combination with once-daily lopinavir/ritonavir (LPV/r), in ART-naïve subjects over 96 weeks[2]**
- **We investigated virologic and immunologic response by sex and racial sub-groups in the HEAT study.**

Methods

- **688 subjects were included in the intent-to-treat-exposed (ITT-E) population from sites across the United States and Puerto Rico.**
- **The primary endpoint was the proportion of subjects with HIV-1 RNA <50 copies/mL (c/mL) at Week 48 by Missing=Failure (M=F) analysis. Subjects that switched off randomized treatment were included (Switch included).**
- **Subjects self-identified race and ethnicity separately. Efficacy outcomes were summarized for all subjects in the ITT(E) population, as well as, by sex (female or male) and by race (african-american, caucasian, hispanic ethnicity or other race).**
- **No formal assessment of medication compliance was performed in this study.**
- **No formal statistical comparisons were performed in this sub-group analysis.**

Results

Table 1. Baseline Demographics of ITT-E Population

Parameter	Total N=688	ABC/3TC + LPV/r n=343	TDF/FTC + LPV/r n=345
Age, (y), median (range)	38 (18-74)	38 (18-74)	38 (18-69)
Sex; n (%)			
Male	82%	287 (84%)	276 (80%)
Female	18%	56 (16%)	69 (20%)
Race[1]; n (%)			
Caucasian	42%	143 (42%)	147 (43%)
African-American	35%	119 (35%)	124 (36%)
Hispanic ethnicity	20%	73 (21%)	62 (18%)
Other[2]	3%	8 (2%)	12 (3%)
CDC Class C; n (%)	16%	55 (16%)	57 (17%)
Hepatitis B positive	4%	19 (6%)	9 (3%)
Hepatitis C positive	7%	27 (8%)	24 (7%)

1. For this race summary, subjects were grouped based on self-reported ethnicity, and for subjects with non-Hispanic ethnicity, their self-identified race were shown.
2. Other represents Asian, American Indian/Alaskan Native, or another race

- **The ITT-E population was balanced between treatment groups. Fewer than 20% of the population was represented by female subjects. However, 49% of the population was represented by non-Caucasian subjects of which 36% were African-American.**

Table 2. Median Baseline Viral Load and CD4+ Count

	ABC/3TC + LPV/r		TDF/FTC + LPV/r	
	BL HIV-1 RNA (n)	BL CD4+	BL HIV-1 RNA (n)	BL CD4+
All Subjects	4.90 (343)	214	4.84 (345)	193
Female	4.72 (56)	213	4.64 (69)	168
Male	4.94 (287)	214	4.89 (276)	200
African-American	4.76 (119)	182	4.85 (124)	137
Caucasian	4.99 (143)	209	4.87 (147)	237
Hispanic	4.91 (73)	247	4.82 (62)	179
Other	4.71 (8)	184	4.45 (12)	232

- **Females had generally lower baseline viral loads in both groups and only African-Americans in the ABC/3TC + LPV/r group compared to the overall population and other sub-groups. CD4+ counts were generally lower at baseline in African-American subjects as well.**

Figure 1. Proportion of Subjects with HIV-1 RNA <50 c/mL at Week 96; ITT(E) Population, M=F Analysis

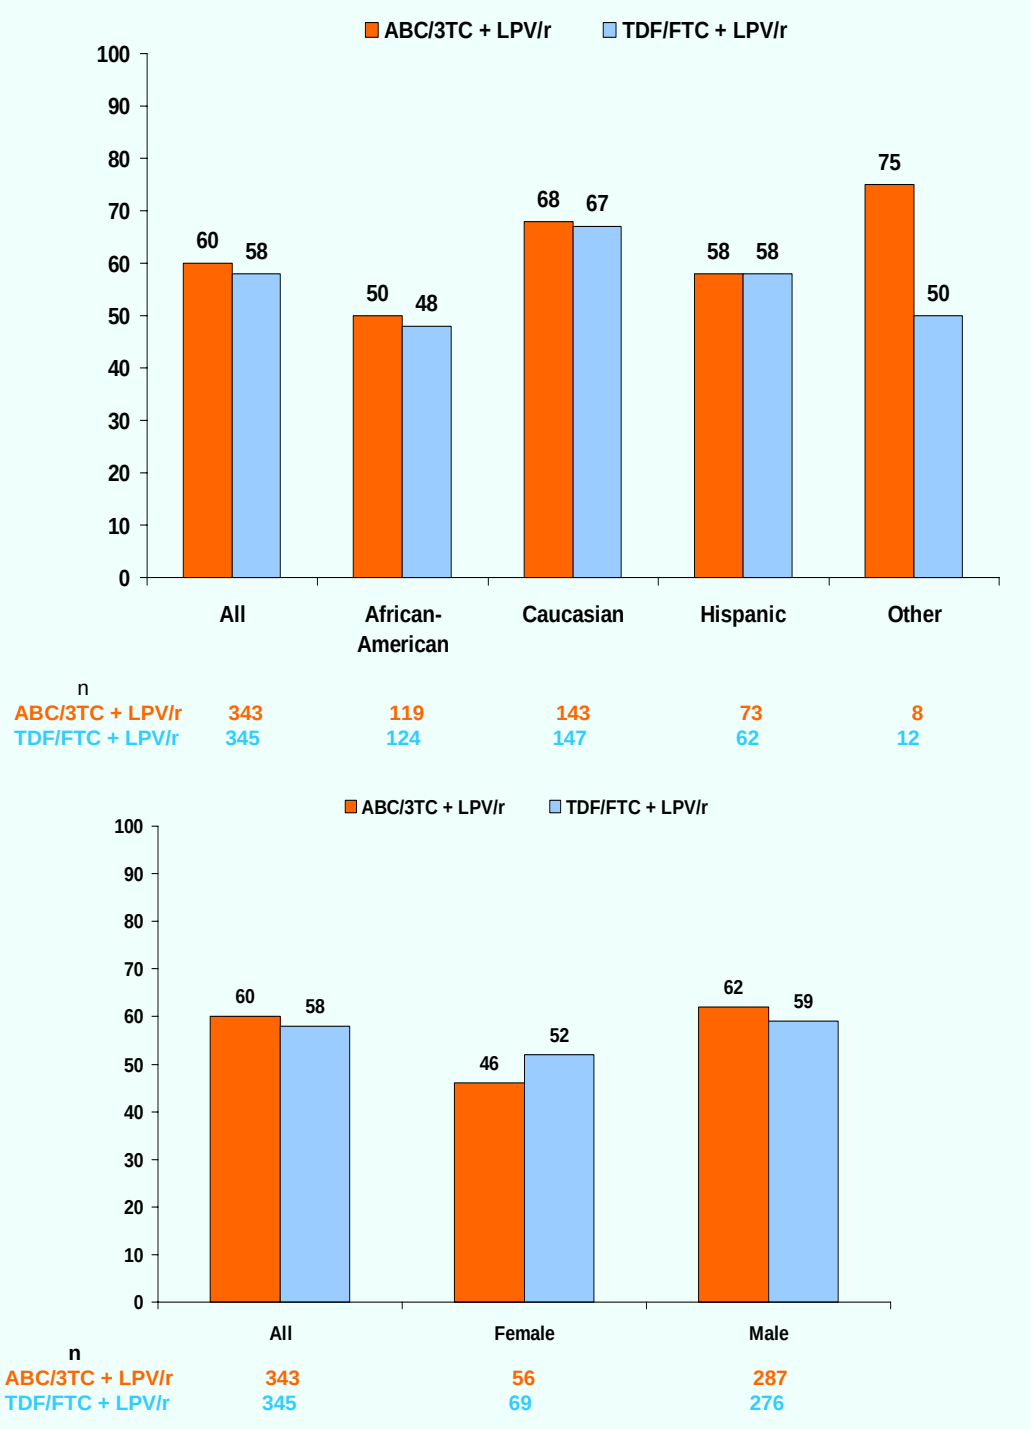
ABC/3TC + LPV/r
TDF/FTC + LPV/r
All 60 58
African-American 50 48
Caucasian 68 67
Hispanic 58 58
Other 75 50
n
ABC/3TC + LPV/r 343 119 143 73 8
TDF/FTC + LPV/r 345 124 147 62 12
All 60 58
Female 46 52
Male 62 59
n
ABC/3TC + LPV/r 343 56 287
TDF/FTC + LPV/r 345 69 276

- **The percentage of subjects with an HIV-1 RNA <50 copies/mL at Week 96 was somewhat lower among female and African-American subjects compared to the overall population and other sub-groups.**

Figure 2. Change from baseline in CD4+ Count at Week 96; ITT(E) Population, Observed Analysis

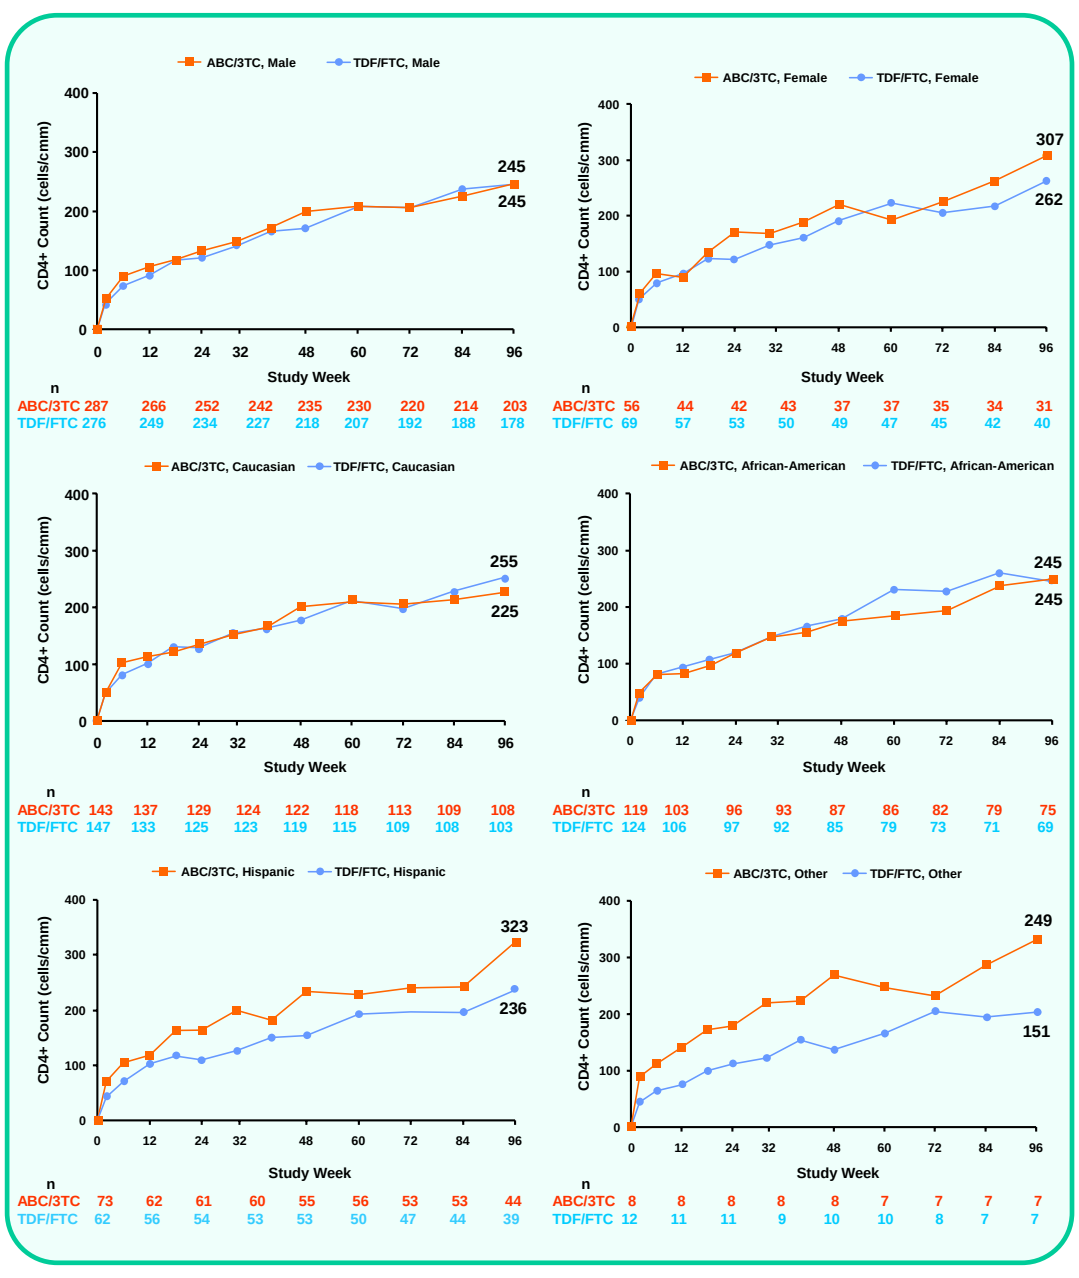
ABC/3TC, Male
TDF/FTC, Male
CD4+ Count (cells/cmm)
Study Week
245
245
n
ABC/3TC 287 266 252 242 235 230 220 214 203
TDF/FTC 276 249 234 227 218 207 192 188 178
ABC/3TC, Female
TDF/FTC, Female
307
262
n
ABC/3TC 56 44 42 43 37 37 35 34 31
TDF/FTC 69 57 53 50 49 47 45 42 40
ABC/3TC, Caucasian
TDF/FTC, Caucasian
255
225
n
ABC/3TC 143 137 129 124 122 118 113 109 108
TDF/FTC 147 133 125 123 119 115 109 108 103
ABC/3TC, African-American
TDF/FTC, African-American
245
245
n
ABC/3TC 119 103 96 93 87 86 82 79 75
TDF/FTC 124 106 97 92 85 79 73 71 69
ABC/3TC, Hispanic
TDF/FTC, Hispanic
323
236
n
ABC/3TC 73 62 61 60 55 56 53 53 44
TDF/FTC 62 56 54 53 53 50 47 44 39
ABC/3TC, Other
TDF/FTC, Other
249
151
n
ABC/3TC 8 8 8 8 8 7 7 7 7
TDF/FTC 12 11 11 9 10 10 8 7 7

- **Overall, female subjects tended to have greater CD4+ recovery compared to males. CD4+ recovery appeared similar between treatment arms within the race and ethnicity sub-groups. Small sample sizes prevented any statistical comparison.**

Discussion

- **Historically, under-representation of women and minorities in clinical trials has hampered our ability to critically examine differences in virologic and immunologic response in these vulnerable subgroups.[3] In the HEAT study enrollment of women was nearly 20% and minorities made up more than 55% of the study population. Thus, this study provides an opportunity to assess treatment responses in these populations.**
- **In this analysis, women appeared to have a reduced virologic response rate compared to the overall population, and this difference was seen in both treatment groups. However, CD4+ cell responses were robust especially in the ABC/3TC group. Despite lower virologic response rates, few women experienced protocol-defined virologic failure.**
- **Fewer African-American subjects achieved virologic suppression compared to the overall population and African-American race was a significant predictor of virologic failure in a multivariate logistic regression model. CD4+ cell responses among African-Americans were not different compared to the overall population.**
- **Reduced treatment responses among women and African-American subjects was also observed in the KLEAN study (FPV/r vs. LPV/r each with ABC/3TC) of 878 treatment-naive subjects through 48 weeks.[4]**

Conclusions

- **Females and African-Americans had reduced treatment response rates compared to the overall and subgroup populations through 96 weeks.**
- **African-American race, but not female sex, was a significant predictor of virologic failure in a multivariate logistic regression model.**
- **Greater CD4+ responses were noted among females in the ABC/3TC group; no treatment differences in CD4+ response were seen among African-Americans in this study.**
- **Large comparative trials of traditionally under-represented populations are needed to further investigate these differences in virologic and immunologic outcomes.**

References

1. Tashima K, Kumar P, Rodriguez-French A, et al. Gender and race subgroup analyses in 4 large, randomized clinical trials comparing abacavir (ABC) to protease inhibitors (PI) or zidovudine (ZDV) in ART-naïve subjects. 8th International Workshop on Adverse Drug Reactions and Lipodystrophy in HIV; 2006 September 24-26; San Francisco, CA, USA.
2. Smith KY, Patel P, Fine D, et al. Randomized, double-blind, placebo-matched, multicenter trial of abacavir/lamivudine or tenofovir/emtricitabine with lopinavir/ritonavir for initial HIV treatment. *AIDS* 2009;published ahead of print, June 23 2009.
3. UNAIDS/WHO. AIDS Epidemic Update, December 2007.
4. DeJesus E, Gathe J, Katlama C, et al. Virologic response and tolerability by sex and race in subjects receiving fosamprenavir/ritonavir (FPV/r) BID and lopinavir/ritonavir (LPV/r) BID, each in combination with abacavir/lamivudine QD (the KLEAN study). 4th International AIDS Society Conference on HIV Pathogenesis, Treatment and Prevention; 2007 July 22-25; Sydney, Australia.

Acknowledgements

The authors would like to extend their thanks to the subjects who participated in this trial and to the study investigators, study coordinators, and the GSK team for their invaluable contributions

IAS 2009- 5th Conference on HIV Pathogenesis, Treatment and Prevention 19-22 July 2009, Cape Town South Africa