Poster Number 485

Efficacy and Safety of Entecavir in Nucleos(t)ide Naïve Asians With HBeAg-Positive and -Negative Chronic Hepatitis B: Results from Studies ETV-022/027

Robert Gish[1], Naoky Tsai[2], Calvin Pan[3], Kris Kowdley[4], Ke-Qin Hu[5], Lai CL[6], Ting-Tsung Chang[7], Kwang-Hyub Han[8], David Cohen[9], Hong Tang[10], Myron Tong[11]

1. Division of Hepatology and Complex GI, California Pacific Medical Center, San Francisco, USA; 2. University of Hawaii, Honolulu, USA; 3. Division of Gastroenterology, Mount Sinai School of Medicine, New York, USA; 4. Center for Liver Disease, Virginia Mason Medical Center, Seattle, USA; 5. Division of Gastroenterology, University of California, Irvine Medical Center, Orange, USA; 6. Queen Mary Hospital, Hong Kong, China; 7. National Cheng Kung University Medical College, Tainan, Taiwan; 8. Severance Hospital, Yonsei University College of Medicine, Seoul, Korea; 9. Research & Development, Bristol-Myers Squibb Company, Wallingford, USA; 10. Research & Development, Bristol-Myers Squibb Company, Plainsboro, USA; 11. Pfleger Liver Institute, University of California School of Medicine, Los Angeles, USA.

BACKGROUND

- Hepatitis B is endemic in China and other parts of Asia
- Entecavir (ETV) 0.5 mg demonstrated superior virologic, histologic and biochemical activity compared to lamivudine (LVD) 100 mg in nucleoside-naïve HBeAg(+) and HBeAg(-) chronic hepatitis B (CHB) patients (Phase III studies ETV-022/027)
- The safety profile of ETV was comparable to LVD in the two studies
- This analysis presents the efficacy and safety data of ETV among Asian patients with CHB participating in the two phase III studies (ETV-022/027) at Week 48

METHODS

Study Population and Design

- CHB patients from Phase III studies ETV-022/027 who:
 - Received ETV 0.5 mg or LVD 100 mg for a minimum of 52 weeks
 - Self-identified as Asian

End Points

- Primary end point was histologic improvement (≥2-point reduction in the Knodell necroinflammatory score, without worsening of the Knodell fibrosis ccore) at Week 48
- Other key end points were virologic, biochemical, and serological response and safety at Week 48

Resistance and Safety Analyses

- Asian HBeAg(+) patients were part of the ETV resistance monitoring program
- Genotyping was performed on paired baseline and on-treatment samples from all patients with:
 - HBV DNA ≥ 300 copies/mL (50 IU/mL) through Week 48
 - Virologic breakthrough (confirmed ≥ 1 $\log_{10}$ increase in HBV DNA from nadir) while on treatment
- Safety was assessed by the incidence of clinical adverse events (AEs) and laboratory abnormalities

STUDY RESULTS

- Nearly half of all patients in ETV-022/027 were Asian:
 - 48% (326/679) on ETV
 - 50% (331/668) on LVD
- Among 657 Asian patients enrolled in the two studies:
 - 406 were HBeAg (+)
 - 251 were HBeAg (-)

Table 1 Demographics and Baseline Characteristics

	ETV-022 (HBeAg[+])		ETV-027 (HBeAg[-])	
	ETV 0.5 mg (N=204)	LVD 100 mg (N=202)	ETV 0.5 mg (N=122)	LVD 100 mg (N=129)
Age, mean (years)	32	32	44	44
Male (%)	158 (77)	140 (69)	97 (80)	110 (85)
HBV DNA by PCR, mean $\log_{10}$ copies/mL	9.64	9.69	7.64	7.35
ALT, mean (U/L)	137	158	144	138
Knodell necroinflammatory Score, mean	7.6	7.6	8.3	7.7
Knodell fibrosis score, mean	1.6	1.5	1.8	1.8
Ishak fibrosis score, mean	2.2	2.1	2.3	2.4
HBV genotype: A, B, C, D, Other	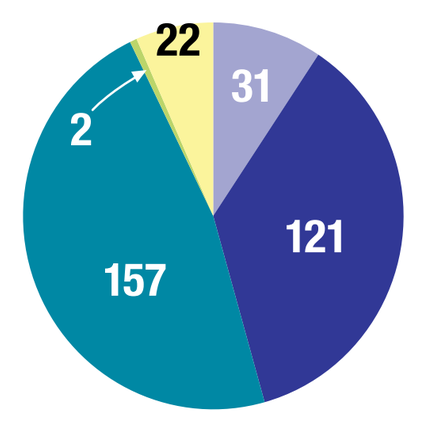 22 31 2 121 157		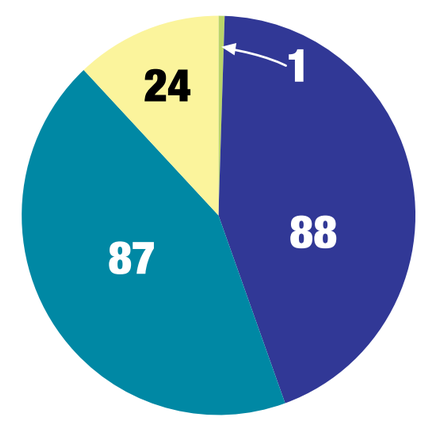 24 1 88 87	

Summary statistics are based on subjects with adequate baseline biopsies

Figure 1 Histologic Improvement Through 48 Weeks

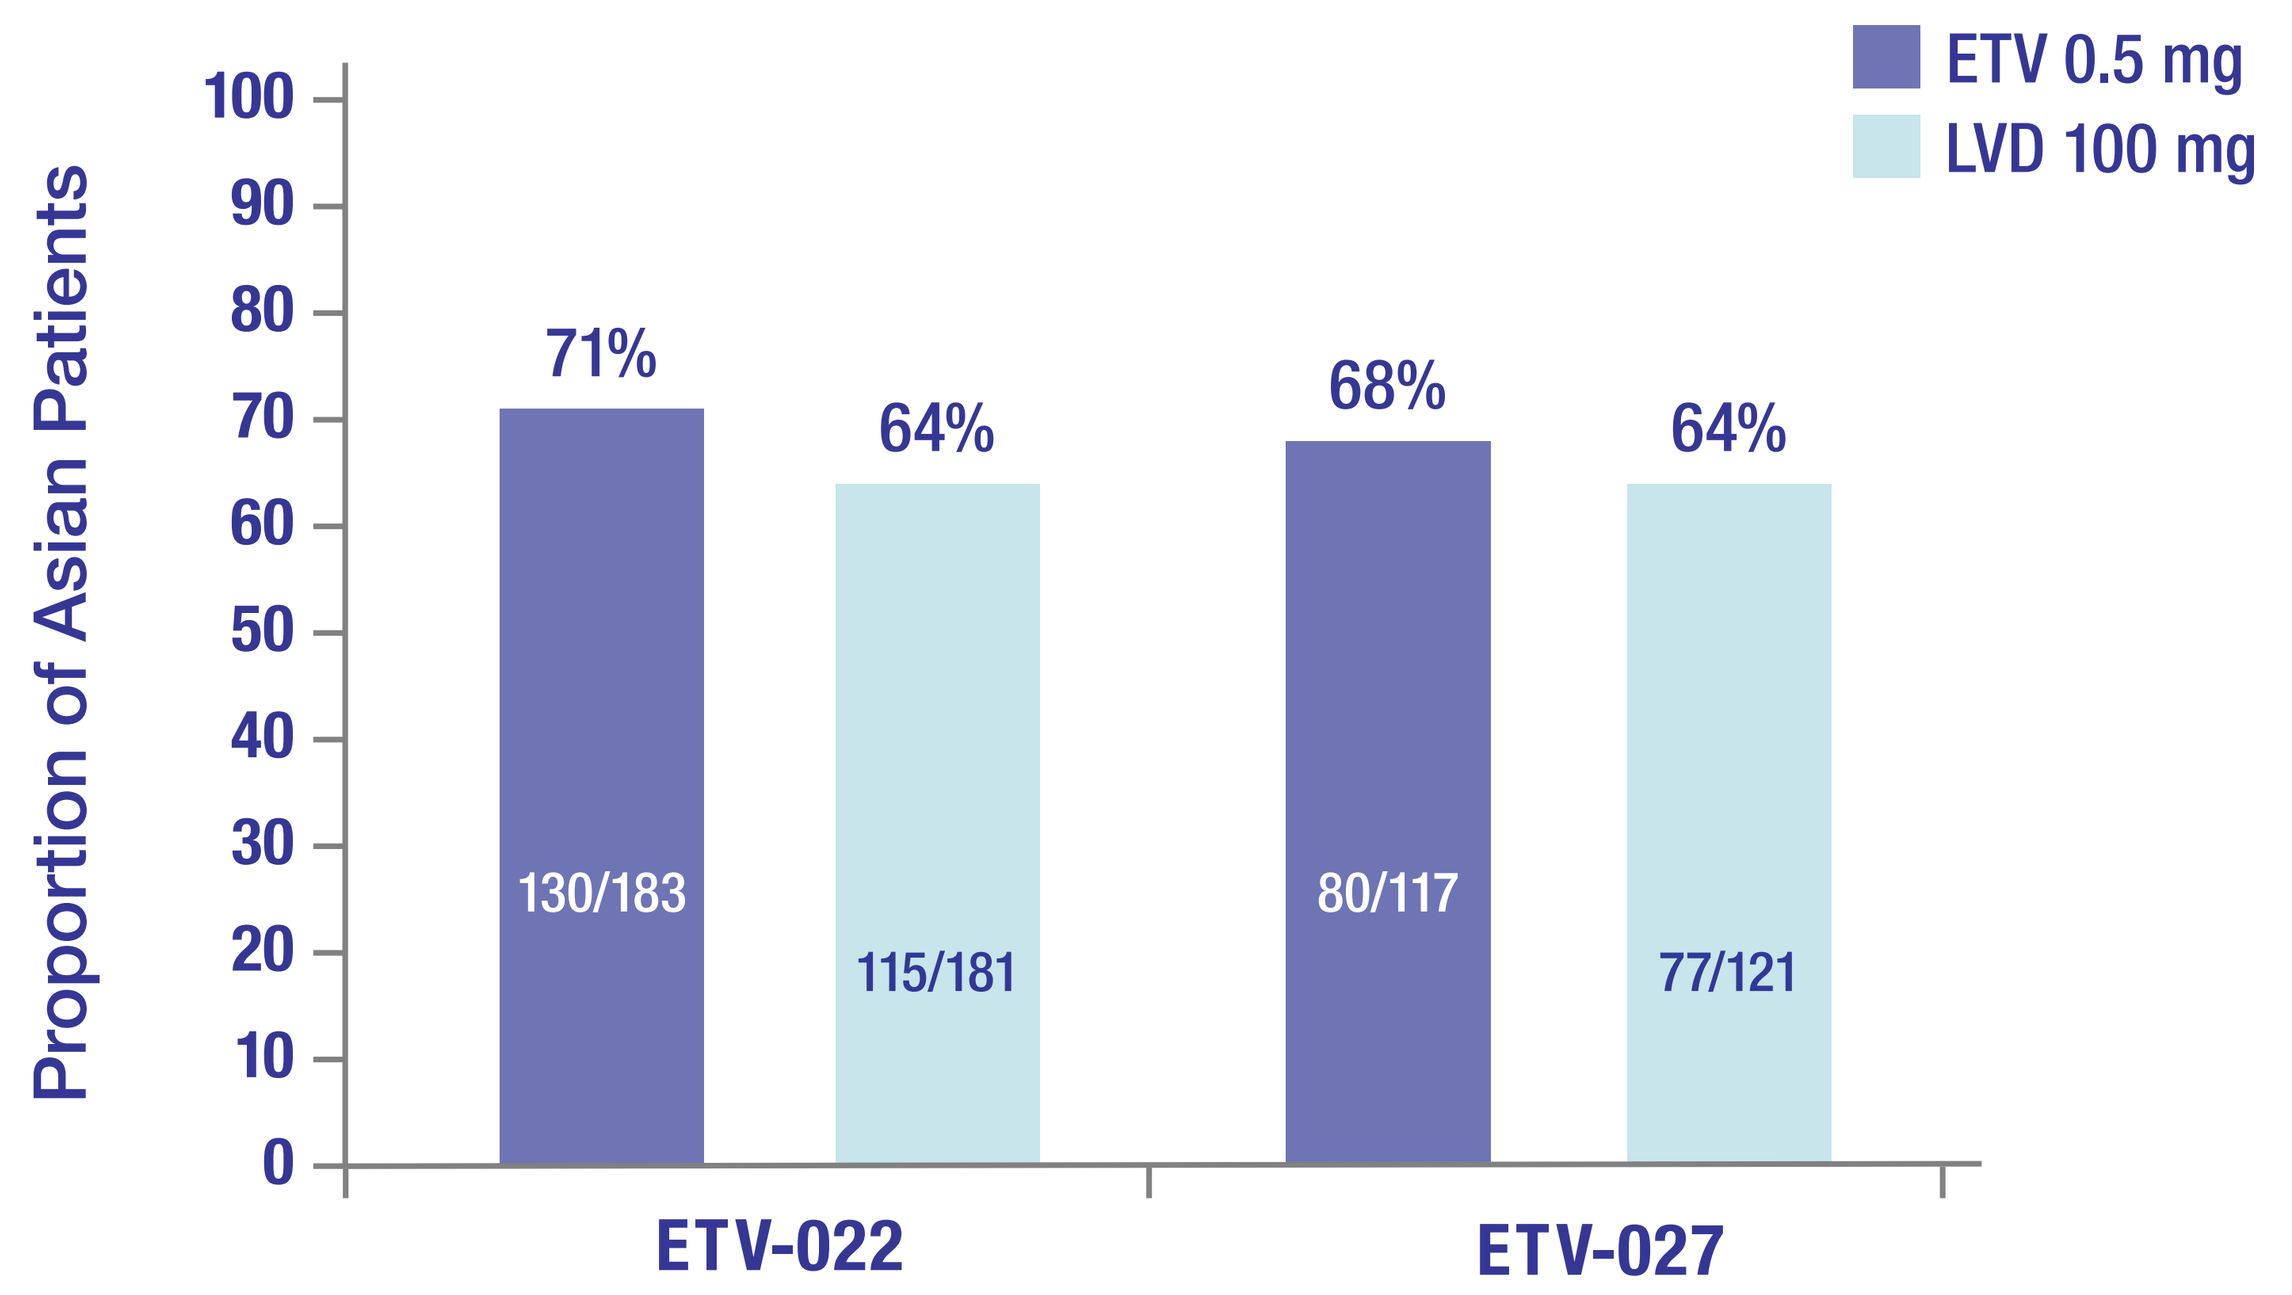
ETV 0.5 mg
LVD 100 mg
Proportion of Asian Patients
100
90
80
70
60
50
40
30
20
10
0
71%
64%
68%
64%
130/183
115/181
80/117
77/121
ETV-022
ETV-027

- Overall, histologic improvement occurred in 70% (210/300) of patients in the ETV group versus 64% (192/302) in the LVD group

Figure 2 Proportion of Asian Patients With HBV DNA <300 copies/mL Through 48 Weeks

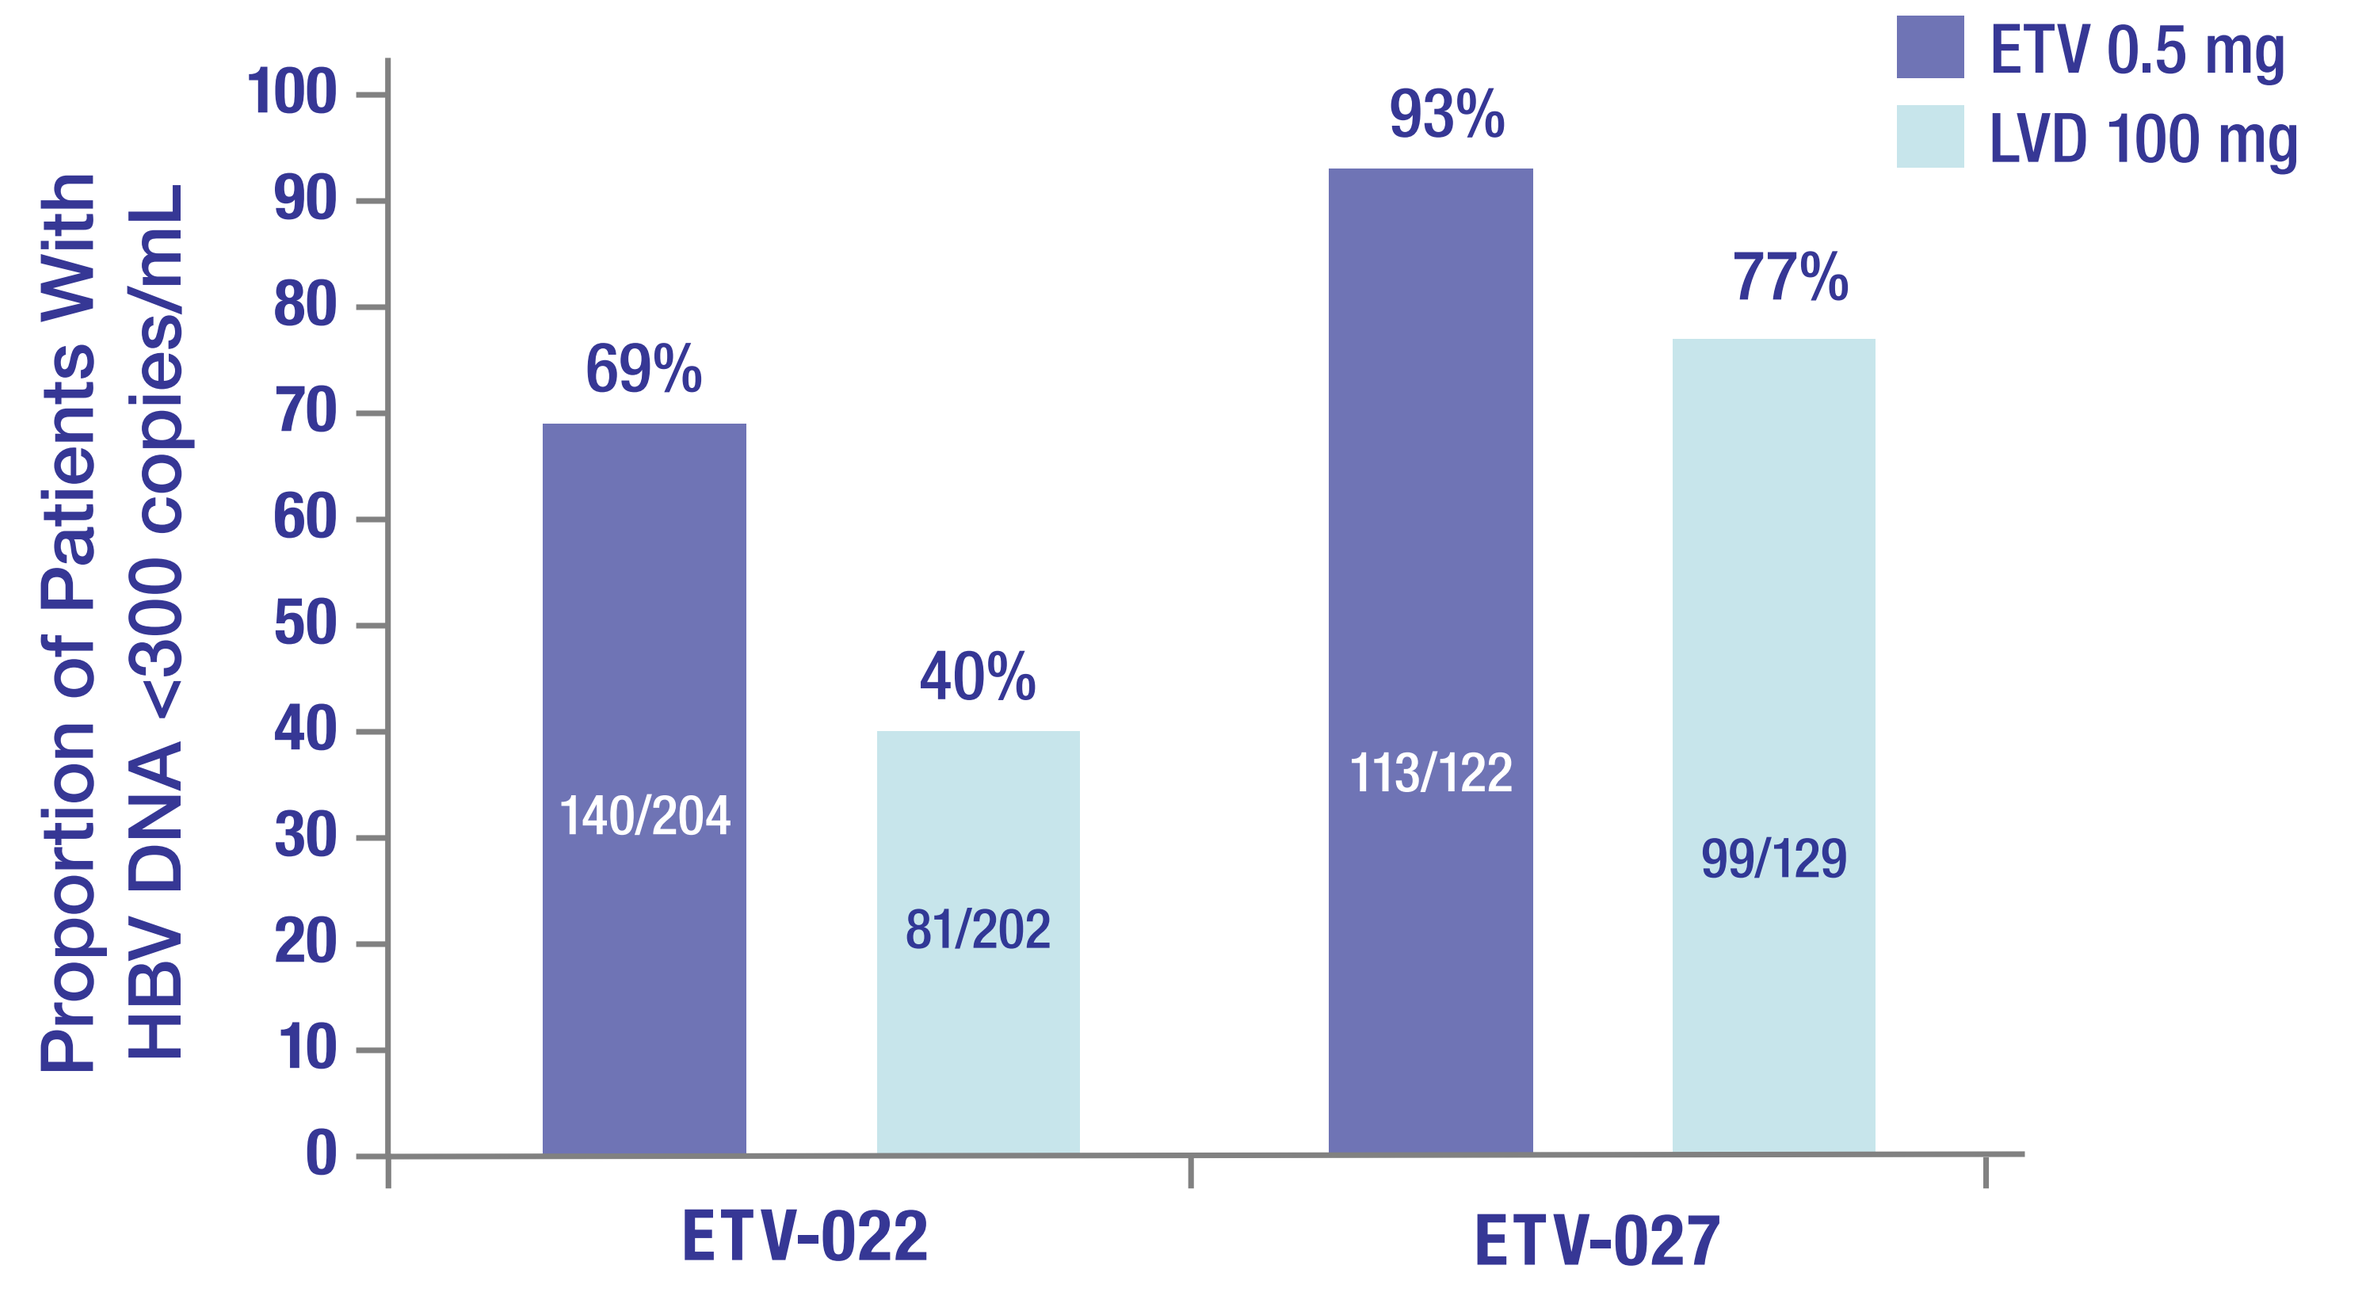
ETV 0.5 mg
LVD 100 mg
Proportion of Patients With HBV DNA <300 copies/mL
100
90
80
70
60
50
40
30
20
10
0
69%
40%
93%
77%
140/204
81/202
113/122
99/129
ETV-022
ETV-027

- Overall, 78% (253/326) of patients in the ETV group and 54% (180/331) in the LVD group had HBV DNA levels <300 copies/mL

Figure 3 Proportion of Asian Patients With ALT ≤1 x ULN Through 48 Weeks

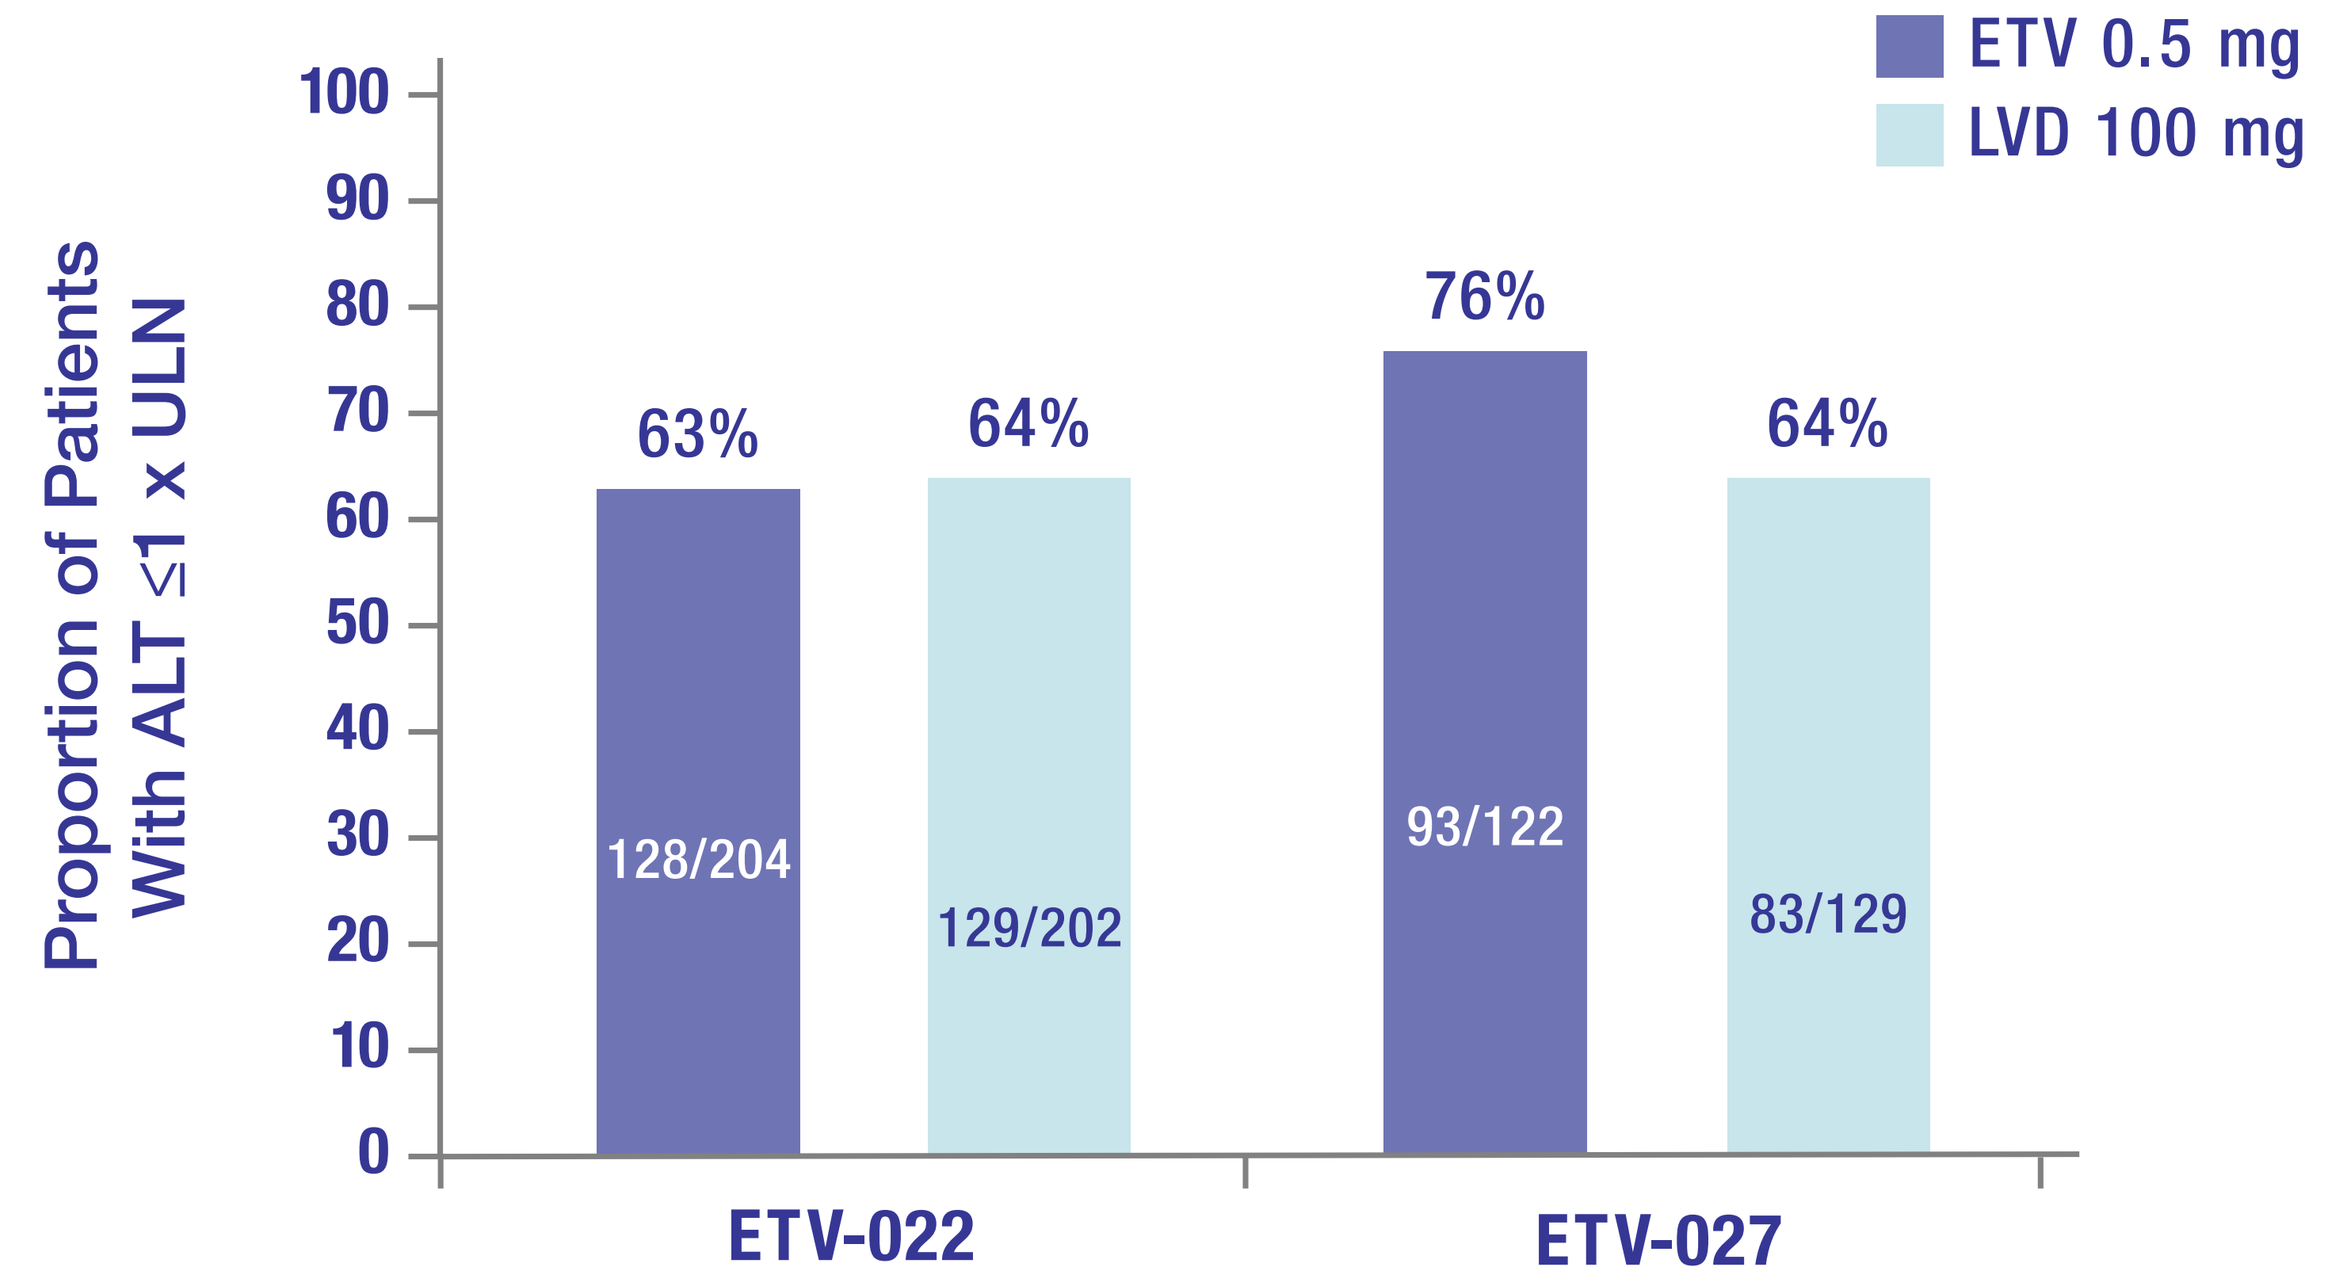
ETV 0.5 mg
LVD 100 mg
Proportion of Patients With ALT ≤1 x ULN
100
90
80
70
60
50
40
30
20
10
0
63%
64%
76%
64%
128/204
129/202
93/122
83/129
ETV-022
ETV-027

- Overall, 68% (221/326) in the ETV group and 64% (212/331) in the LVD group had ALT normalization

Serologic Response in HBeAg(+) Asian Patients Treated With ETV Through Week 48

- 16% (33/204) of patients had loss of HBeAg
- 16% (32/204) of patients had HBeAg seroconversion
- One (1) patient had HBsAg loss

Resistance in Both HBeAg(+) and HBeAg(-) Asian Patients Treated With ETV Through Week 48

- No Asian patients developed ETV resistance

Table 2 Cumulative Safety of Asian Patients from ETV-022 and ETV-027

	ETV 0.5 mg (N=326) N (%)	LVD 100 mg (N=331) N (%)
Any adverse event	287 (88)	283 (85)
Grade 3-4 adverse events	31 (10)	34 (10)
Serious adverse events	18 (6)	21 (6)
Discontinuation due to adverse events	3 (<1)	7 (2)
All deaths	1* (<1)	0 (<1)
On-treatment ALT flare†	7 (2)	7 (2)

* There was one death in the ETV 0.5 mg arm in study ETV-027 on Day 314 due to liver cancer

† ALT flare: ALT >2 x baseline and >10 x ULN by clinical laboratory evaluation

SUMMARY OF RESULTS

- In this subanalysis of Asian patients enrolled in studies ETV-022/027, ETV use at Week 48 demonstrated:
 - Histologic improvement in 70% of patients
 - HBV DNA <300 copies/mL in 78% of patients
 - ALT normalization in 68% of patients with HBeAg loss and seroconversion occurring in 16% of HBeAg(+) patients
- Resistance did not develop in any Asian patient in ETV-022/027
- Safety was comparable to previously reported data

CONCLUSIONS

- ETV 0.5 mg demonstrated significant histologic, virologic, and biochemical responses compared with LVD 100 mg in nucleoside-naïve Asian patients with HBeAg positive and negative chronic hepatitis B at Week 48
- Efficacy and safety profiles for ETV were similar and consistent with the findings in the overall studies

Disclosures

- David Cohen and Hong Tang are employees of Bristol-Myers Squibb
- Robert Gish, Naoky Tsai, Calvin Pan, Kris Kowdley, Ke-Qin Hu, Lai CL, Ting-Tsung Chang, Kwang-Hyub Han, and Myron Tong have received research support from Bristol-Myers Squibb

61st Annual Meeting of the American Association for the Study of Liver Diseases. Presented October 29 – Novermber 2, 2010

Bristol-Myers Squibb