Chronic Hepatitis C (HCV) Infections and the Risk of Depression and Other Adverse Events

Jeffrey McCombs, PhD[1], Yong Yuan, PhD[2], Janet Shin, PharmD[1], Sammy Saab, MD[3]

[1]USC School of Pharmacy, Los Angeles, CA, USA; [2]Bristol Myers-Squibb, Princeton, NJ, USA; [3]David Geffen School of Medicine, UCLA, Los Angeles, CA, USA

BACKGROUND

Hepatitis C (HCV) is a major medical and public health concern worldwide. In the United States, the Centers for Disease Control estimate the prevalence of chronic HCV at between 2.7 and 3.9 million Americans.[1] The cost associated with chronic HCV infection in the U.S. was estimated at $1.8 billion in 1997 and these costs are expected to increase significantly over the next 2 decades.[2,3] Serious complications of HCV may develop, such as liver failure, liver transplantation and cancer.

HCV patients may suffer from depression which is thought to be associated with treatment. However, a rigorous study design is required to measure the incremental effects of HCV on the risk of depression and other adverse clinical outcomes. Specifically, HCV patients must be compared to similar patients without an HCV diagnosis and multi-variable statistical methods are required to adjust for any remaining potential confounding factors.

OBJECTIVES

Estimate the incremental impact of HCV on incidence of depression and other serious clinical events adjusting for age, gender, and non-HCV related co-morbidities.

METHODS

Data for this study were derived from a large health insurance company in the U.S. These data consisted of all paid claims for all enrollees covered during a 6 year period [2003 to 2008]. The study then selected a sample of enrollees with a minimum of two years of uninterrupted eligibility.

Patients with an HCV infection were identified based on ICD-9 diagnostic data recorded on paid claims [070.41, 070.44, 070.51, 070.54, 070.70, 070.71, or V02.62]. A two year observation period was defined for each HCV patient centered on the first observed HCV diagnosis or the start of drug therapy for HCV [index date]. A corresponding 2 year observation period was defined for all non-HCV patients based on their first day of eligibility and their index date was set at one year following their eligibility date. Non-HCV patients were matched to HCV patients on a 1-to-1 ratio using a propensity score derived from a parsimonious logistic regression model of the likelihood of an HCV infection. The list of potential risk factors included age, gender, type of insurance [Medicare, Medicaid, commercial], plan type [e.g., HMO, indemnity, PPO, etc.], state of residence, baseline diagnostic mix, prescription drug profile, and the prior use of health services.

Finally, HCV and non-HCV patients with hepatitis B, HIV, cirrhosis, liver cancer or a liver transplant during the year prior to their index date were excluded from the analysis.

The risk of liver-related clinical events was estimated by comparing the matched HCV and non-HCV patients. Adverse events were identified based on the filing of a paid claim with one of the following diagnoses: liver cancer [155.0]; liver transplantation [996.82, V42.7, CPT codes: 00796, 47135, 47136], depression [296.2, 296.3, 300.4, 311] and cirrhosis [571.5, 571.2, 571.6]. Differences in event rates between the HCV and matched non-HCV populations were documented using descriptive statistics and chi square statistics were calculated. Multivariate models of adverse events [logistic regression] were estimated which included all baseline demographic, diagnostic, and drug profile variables listed above.

RESULTS

A total of 60,806 HCV patients were identified using the 6 years of paid claims data based on two or more HCV related diagnoses or a single diagnosis combined with a prescription for ribavirin, interferon-alpha, pegylated interferon-alpha or interferon-alfcon. Of this HCV population, only 8,861 patients [14.6%] exhibited a full year of eligibility prior to their index date without any evidence of cirrhosis, liver cancer or liver transplantation. An additional 410,605 non-HCV patients were identified using the 6 years of paid claims data who also had a minimum of 2 years of continuous eligibility and no evidence of HCV related adverse events in the pre-index year. These 419,466 patients were used to estimate logistic models of the factors associated with HCV infection. The factors found to be significantly associated with HCV are summarized in Figures 1-4.

Figure 1: Association between Age and the Presence of a reported HCV diagnosis in the general population

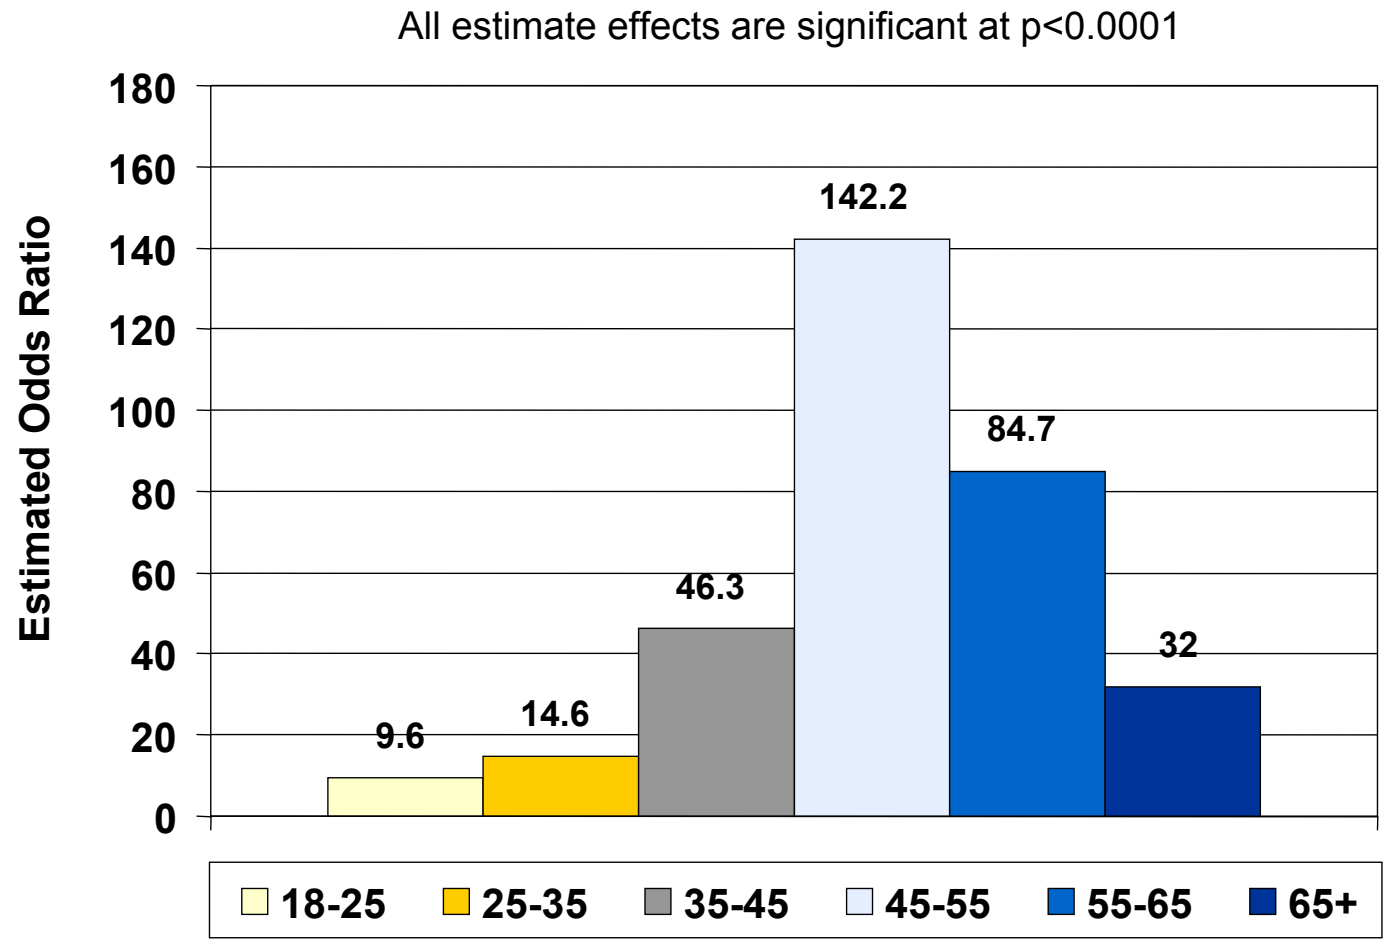
All estimate effects are significant at p<0.0001
Estimated Odds Ratio
180
160
140
120
100
80
60
40
20
0
9.6
14.6
46.3
142.2
84.7
32
18-25
25-35
35-45
45-55
55-65
65+

Figure 2: Co-Morbidities Associated with a Reported HCV Diagnosis in the general population

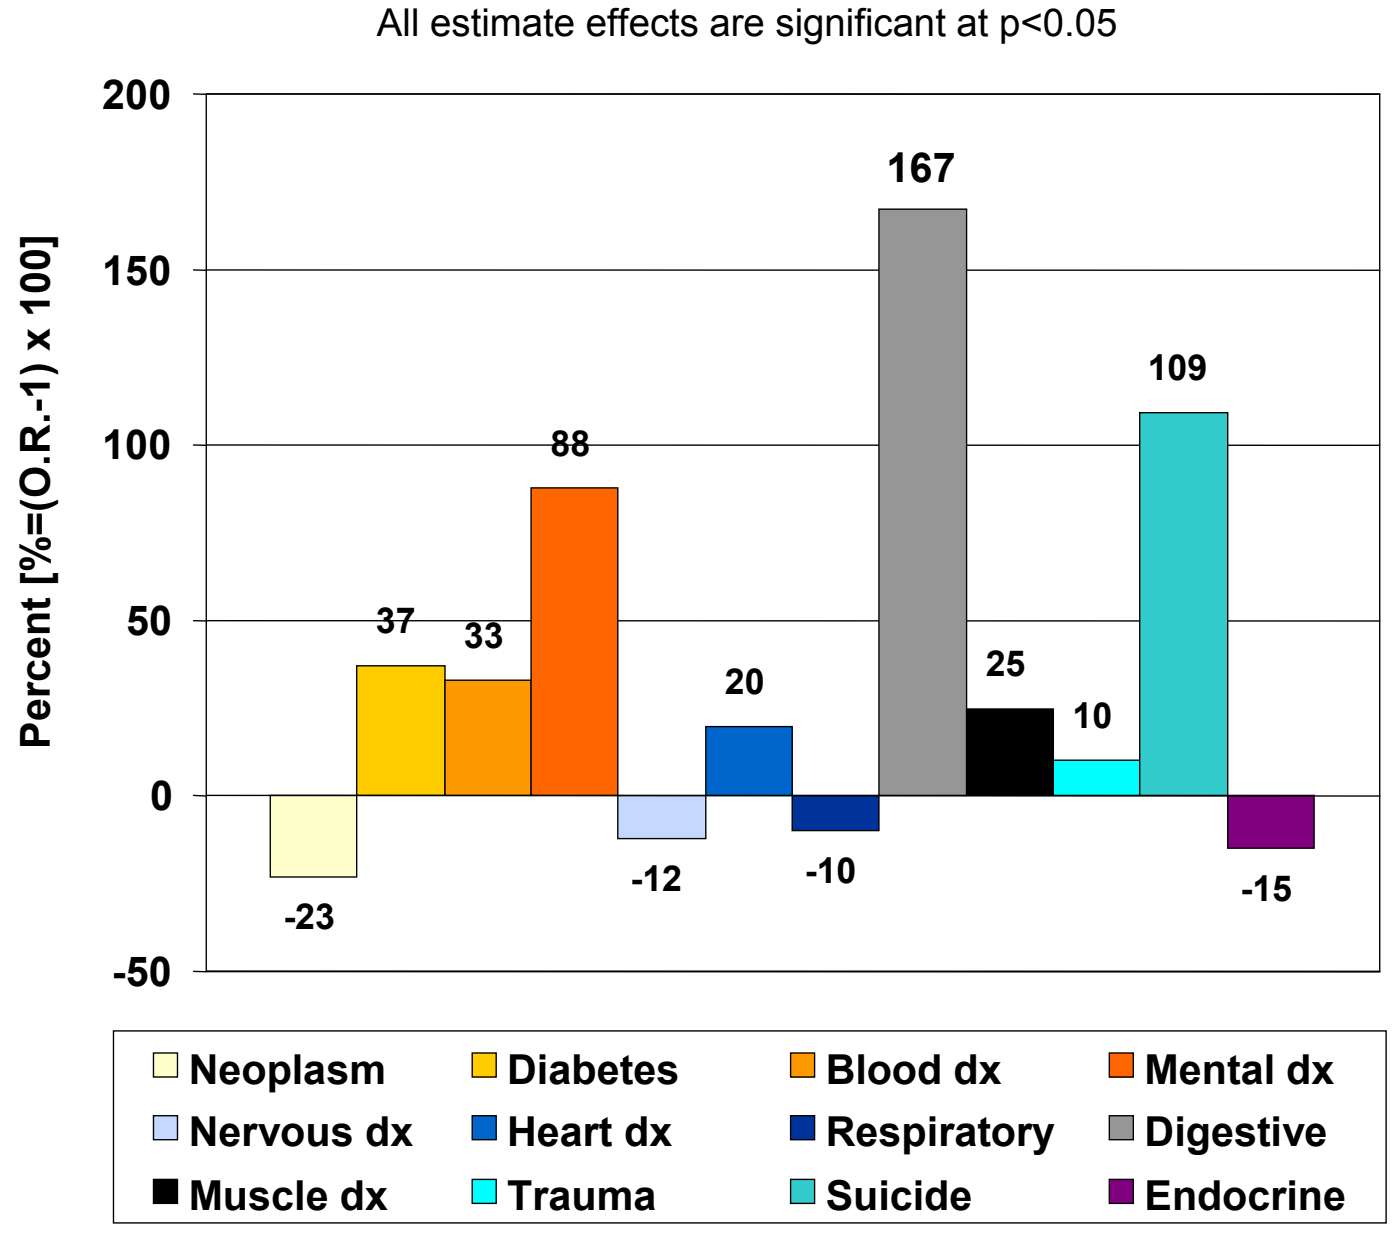
All estimate effects are significant at p<0.05
Percent [%=(O.R.-1) x 100]
200
150
100
50
0
-50
-23
37
33
88
-12
20
-10
167
25
10
109
-15
Neoplasm
Diabetes
Blood dx
Mental dx
Nervous dx
Heart dx
Respiratory
Digestive
Muscle dx
Trauma
Suicide
Endocrine

RESULTS (cont'd)

Figure 3: Other Patient Characteristics Associated with a Reported HCV Diagnosis in the General Population

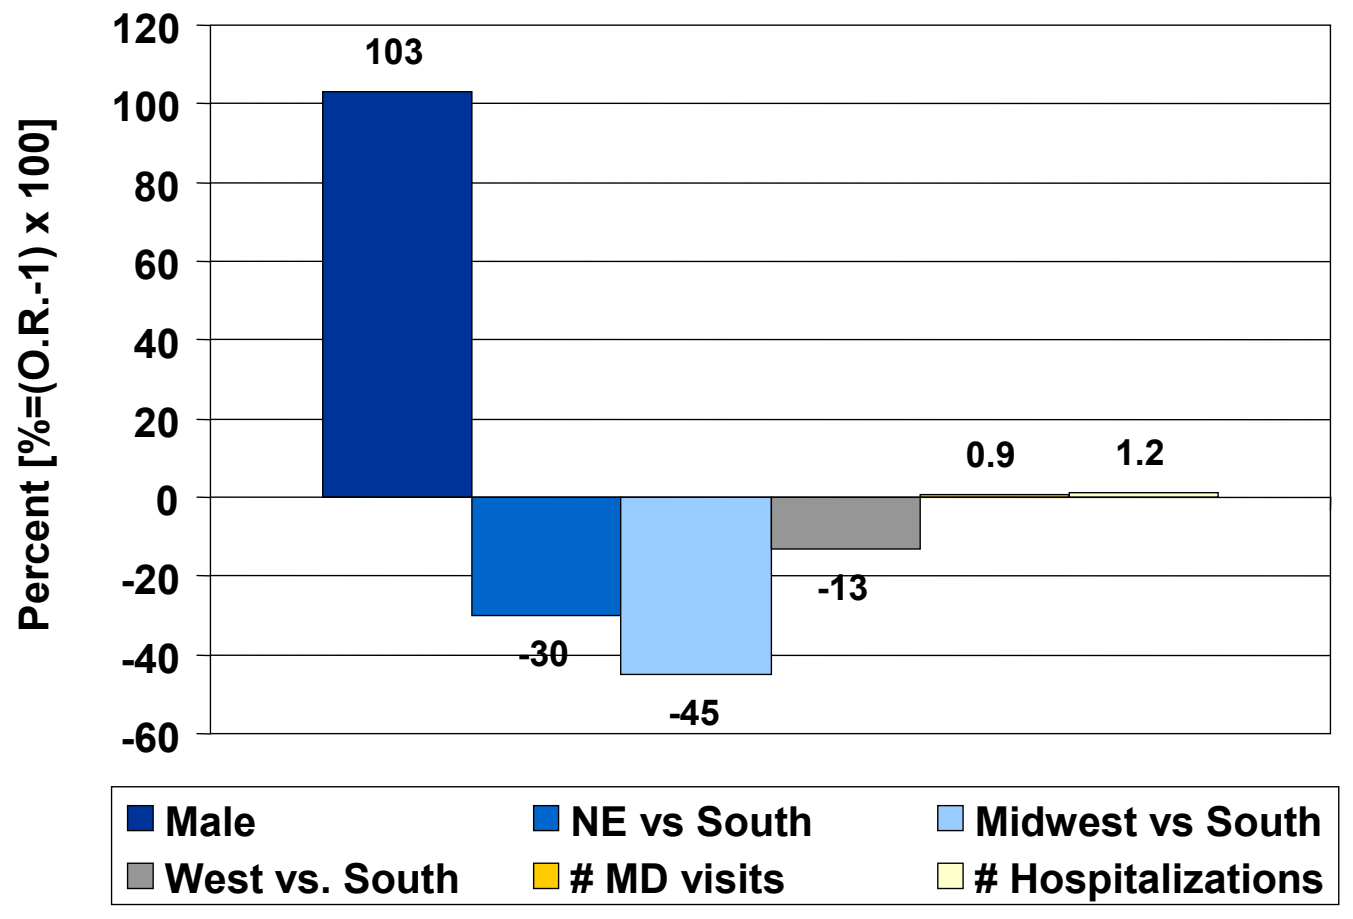
Percent [%=(O.R.-1) x 100]
120
100
80
60
40
20
0
-20
-40
-60
103
-30
-45
-13
0.9
1.2
Male
NE vs South
Midwest vs South
West vs. South
MD visits
Hospitalizations

The results from the propensity score model of the risk factors associated with HCV are consistent with the literature. Patients aged 35 and above are at much greater risk than younger patients, especially patients aged 45-55 on their index date during the period 2003-2008. Males are twice as likely as females to be infected with HCV and the risk is much higher for patients living in the south relative to the west, mid-west or north east.

The results of the propensity score model were used to match 8,861 HCV patients with 8,861 non-HCV patients. The matched HCV and non-HCV patients did not differ significantly on any pre-index characteristics including demographic characteristics, health insurance coverage, prior use of services, health care cost, prescription drug profile or co-morbidity profile [data not presented].

The simple counts of adverse events in the post-index year are presented in Figure 4. Estimates of the incremental impact of HCV on event risk were estimated using logistic regression models and these results are compared to the unadjusted odds ratios based on counts in Figure 5. The rate of depression in the first year is presented in Figure 6 broken down by treatment status documenting higher rates of depression in treated HCV patients.

Figure 4: Count of Adverse Events in First Post-Index Year

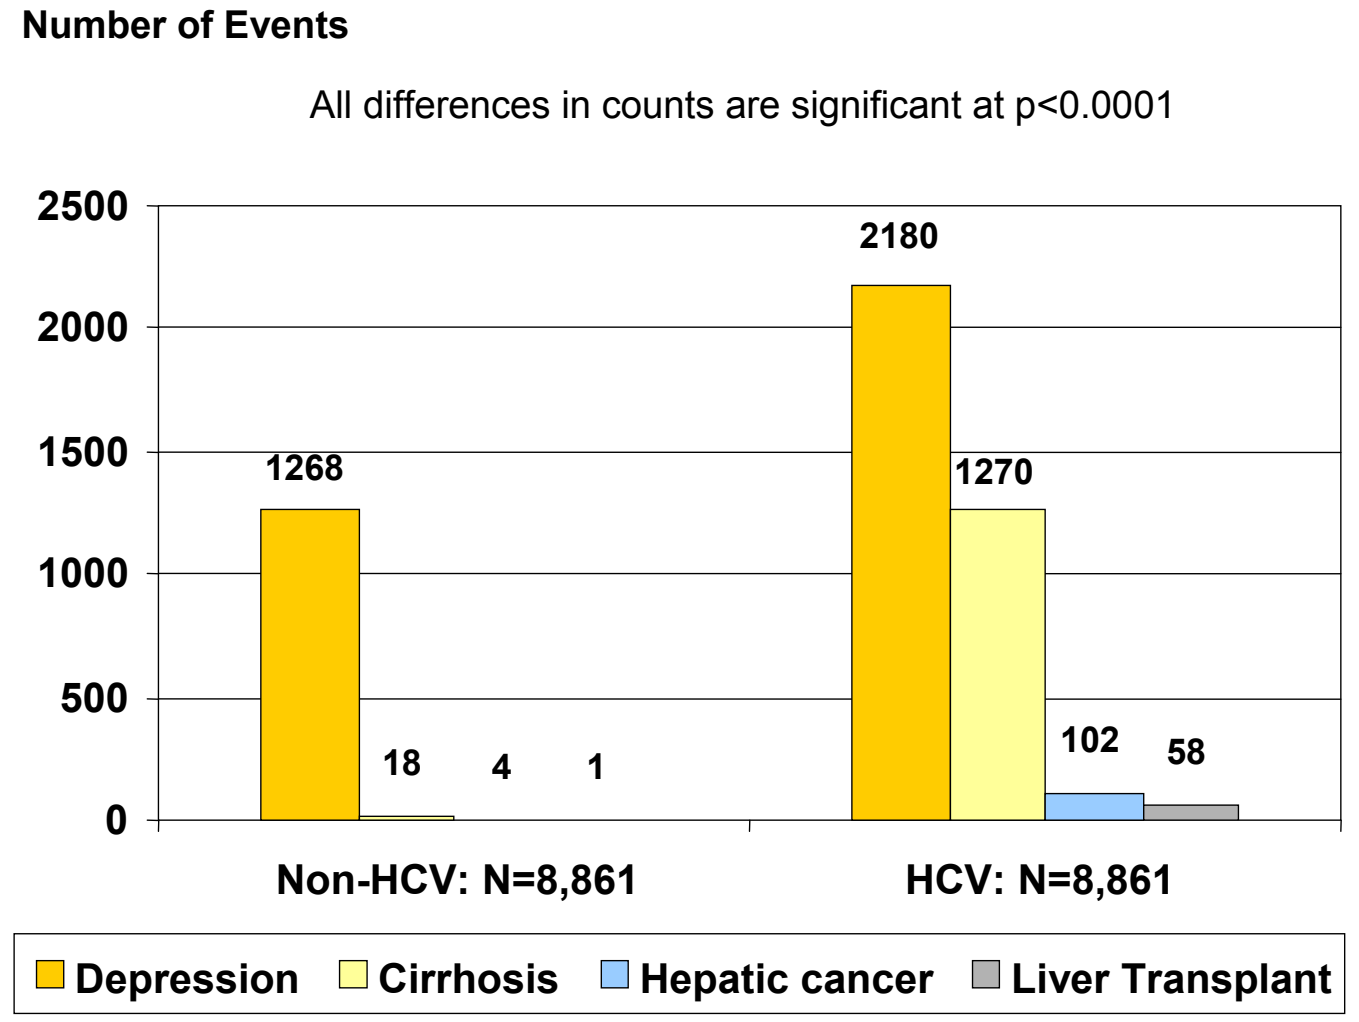
Number of Events
All differences in counts are significant at p<0.0001
2500
2000
1500
1000
500
0
1268
18
4
1
2180
1270
102
58
Non-HCV: N=8,861
HCV: N=8,861
Depression
Cirrhosis
Hepatic cancer
Liver Transplant

RESULTS (cont'd)

Figure 5: Incremental Effects of HCV on Event Risk [Unadjusted vs Adjusted]

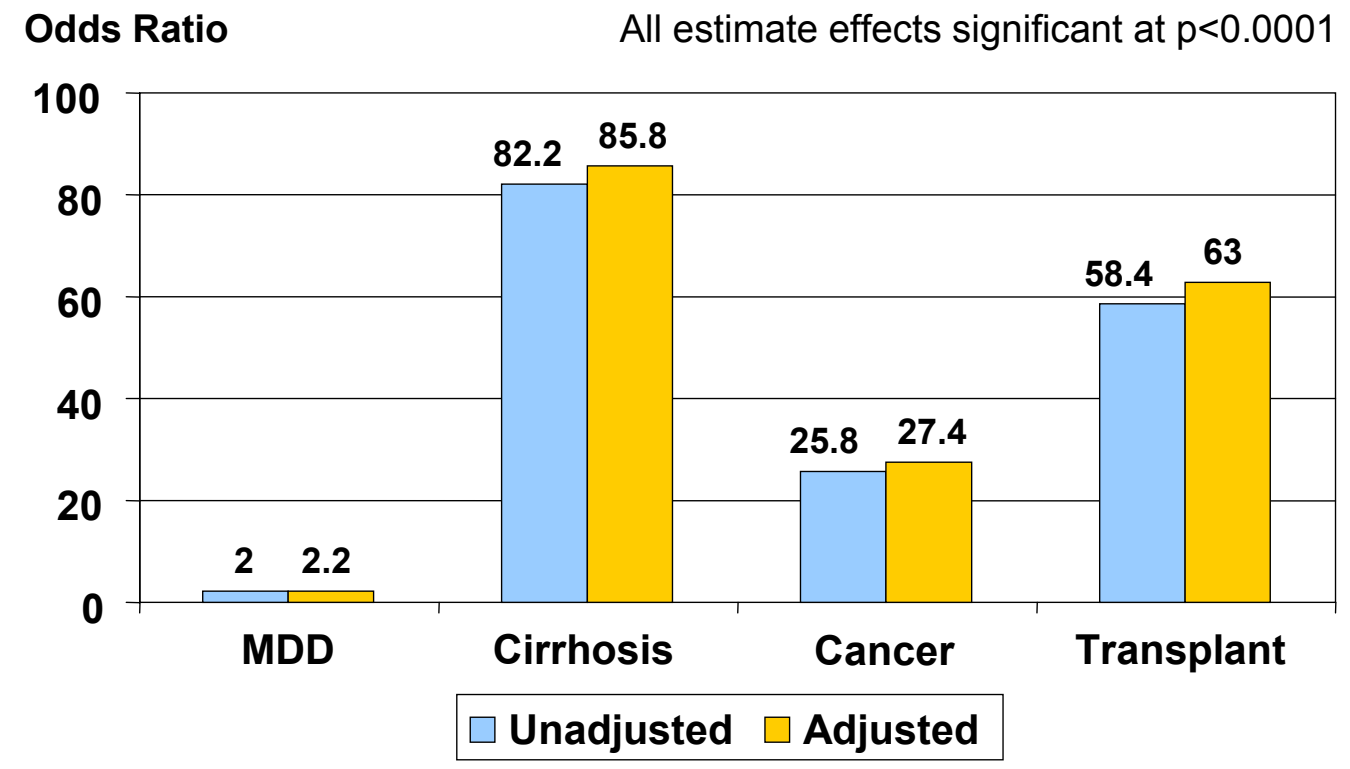
Odds Ratio
All estimate effects significant at p<0.0001
100
80
60
40
20
0
2
2.2
82.2
85.8
25.8
27.4
58.4
63
MDD
Cirrhosis
Cancer
Transplant
Unadjusted
Adjusted

Figure 6: Rate of Depression in First Post-Index Year

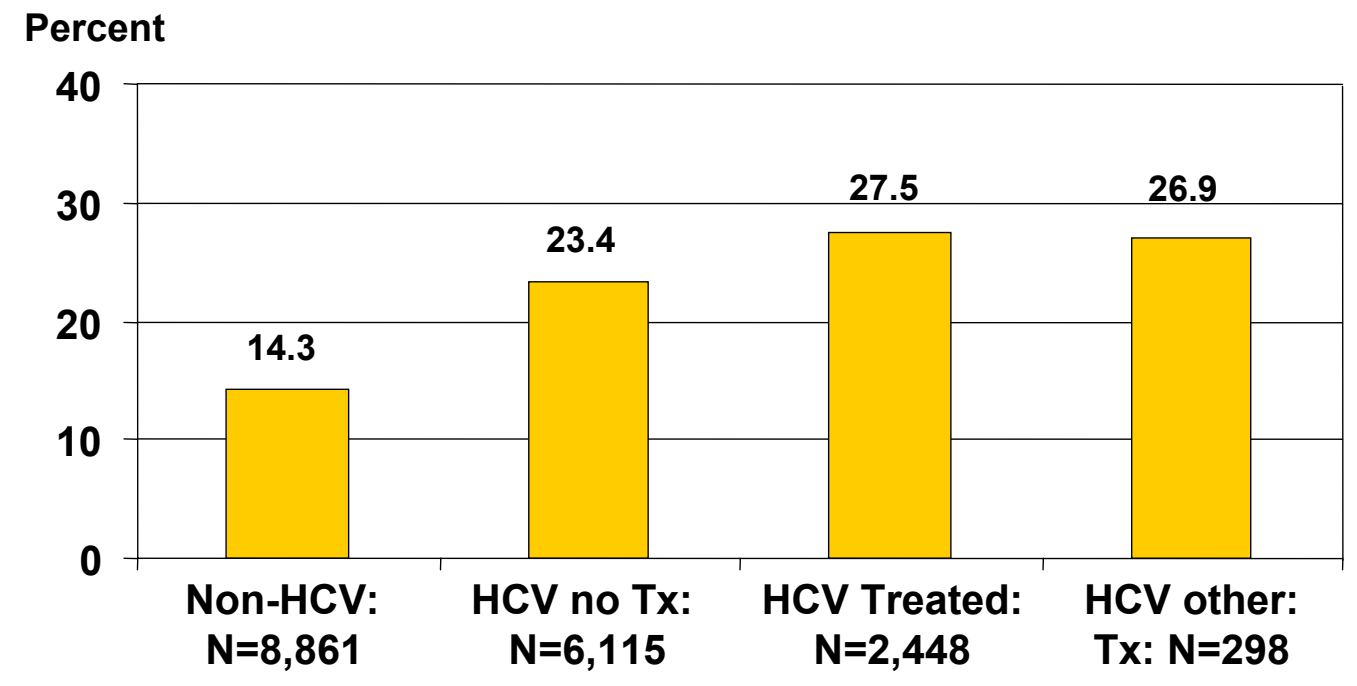
Percent
40
30
20
10
0
14.3
23.4
27.5
26.9
Non-HCV: N=8,861
HCV no Tx: N=6,115
HCV Treated: N=2,448
HCV other: Tx: N=298

HCV SOC = pegylated interferon-alpha + ribavirin; HCV Other Tx = all other combinations of HCV related therapies

CONCLUSIONS

This study estimates the extent to which HCV significantly increases the risk of adverse clinical events by comparing matched HCV and non-HCV populations. HCV doubles the risk of depression and increases the risk of cirrhosis by 80-fold. The likelihood of liver transplantation was increased by a factor of over 60, while the likelihood of developing liver cancer increased 25-fold. Odds ratio data can be misleading if the absolute risk of the event is very low. This is not the case here.

In the year following the diagnosis of HCV in patients without known cirrhosis, nearly 25% of HCV patients experienced depression and 14% had a cirrhosis diagnosis reported. The incidence rate for hepatic cell cancer was over 1 per hundred HCV patients in the first year compared to 5 per 10,000 in the matched, non-HCV population. Liver transplantation remains relatively uncommon in the HCV sample at 58 transplants per 10,000. In a sensitivity analysis [Figure 6], we found that treated HCV patients had higher rates of depression than un-treated HCV patients during the first year.

These results highlight the need for safe, tolerable and effective antiviral therapies. With new therapies under development, there is a need for data on the incremental impact of HCV on the risk of hepatic events and depression. The data presented here can be used in future evaluations of the cost effectiveness of new therapies.

REFERENCES

1. Centers for Disease Control and Prevention, Viral hepatitis: Statistics and surveillance. 2009 http://www.cdc.gov/hepatitis/Statistics.htm.
2. Leigh JP, et al. Cost of hepatitis C. *Archives of Internal Medicine* 2001;161(18):2231-2237.
3. Wong J, McQuillan G, McHutchison J and Poynard T. Estimating future hepatitis C morbidity, mortality, and costs in the United States. *AJPH* 2000;90(10):1562-1569.

This research was funded as an unrestricted grant from Bristol Myers-Squibb to USC which retains all rights for publication of study results

AASLD, The Liver Meeting in Boston, MA, October 29 – November 2, 2010

MedPos10_0149A